221

First-Year Incidence of Adenovirus Infections Following Allogeneic Hematopoietic Cell Transplants in a Pediatric Population

Essy Mozaffari,[1] PharmD, MPH, MBA; Lee Stern,[2] MS; Yi-Chien Lee,[2] MS; Vishal Patel,[2] BS; Roman Casciano,[2] MSc

[1]Chimerix, Durham, NC; [2]LASER Analytica, New York, NY

Presenting author contact details
E Mozaffari
Chimerix Inc.
2505 Meridian Parkway, Suite 340
Durham, NC 27713
Tel: (919) 806-1074
Fax: (919) 806-1146
email: emozaffari@chimerix.com

INTRODUCTION

- Disseminated adenovirus (AdV) infection is associated with high mortality rates in the pediatric population who have undergone hematopoietic cell transplantation (HCT).[1]
- The incidence and predictors of disseminated AdV are not well described.
- Our objective was to describe the first-year incidence of AdV infections in pediatric recipients of allogeneic HCT (allo-HCT) enrolled in a US health insurance plan.

METHODS

Data source

- Data for this study were extracted from the MarketScan Research Databases, which contain healthcare insurance claim records for commercial and Medicare patients.

Patients

- Patients were included in the study if they had a relevant International Classification of Diseases, Ninth Revision (ICD-9) or Current Procedural Terminology procedure code for an allo-HCT between June 26, 2010, and June 30, 2014.
- Patients who were less than 20 years old (pediatric population) at the time of transplant were included in the study.
- Patients must have had at least 360 days of health insurance plan enrollment prior to the allo-HCT procedure.
- The first admission for an allo-HCT procedure for each patient was defined as the index procedure.

Baseline characteristics

- Demographic and clinical characteristics reported in the MarketScan Research Databases were compiled for the study population, including:
 - age at time of transplant
 - sex
 - insurance type
 - region
 - underlying conditions for which the transplant was performed.

Outcome measures

- Patients with AdV infection were identified as those with at least one ICD-9 diagnosis code of AdV infection documented in the health insurance claims records within 365 days following transplant.
- Among those patients with evidence of AdV infection, the setting of the infection diagnosis was documented as occurring:
 - In the initial hospital admission, when the first AdV infection diagnosis was associated with the initial transplant admission.
 - In a subsequent hospital admission, when the first AdV infection diagnosis was associated with a readmission after discharge from the initial transplant admission.
 - In any other place of service when the first ICD-9 diagnosis codes for AdV infection were not associated with their initial transplant admission nor a subsequent hospital admission (i.e., a non-hospital setting).
- Time to first AdV infection diagnosis (days) was reported for patients with evidence of AdV infection.

Statistical analysis

- Mean (standard deviation [SD]) or n (%) were reported for baseline characteristics.
- AdV infection:
 - Number and percentage of patients with at least one ICD-9 code for AdV infection.
 - Incidence rate for AdV infection defined as the number of infections divided by total patient-years of follow-up.
 - – Follow-up time is defined as time from day of transplant up to 365 days or the end of health insurance plan enrollment (whichever was earlier).
 - Setting of AdV infection diagnosis:
 - – number and percentage of patients in each setting were reported.
 - Time to first AdV infection diagnosis (days):
 - – mean (SD), median, first quartile (Q1) and third quartile (Q3) were reported.

RESULTS

Study population

- 350 pediatric allo-HCT recipients were identified within the MarketScan Research Databases as meeting the inclusion criteria, with a mean age of 10.2 (SD=5.4) years; 205 (58.6%) were male (**Table 1**).
 - Most patients (n=248, 70.9%) were on a commercial Preferred Provider Organization/Point-of-Service (PPO/POS)-type insurance plan.
 - Most patients lived in the southern region of the US (n=117, 33.4%).
 - The majority of patients had one or two pre-specified underlying conditions, with the most commonly identified conditions being aplastic anemia (n=150, 42.9%) and acute lymphoblastic leukemia (n=114, 32.6%) (**Table 1**).

Table 1. Characteristics of study population

Characteristics		Pediatric allo-HCT recipients (n=350)
Age (years), mean (SD)		10.2 (5.4)
Male, n (%)		205 (58.6)
Insurance plan type, n (%)	Commercial PPO/POS	248 (70.9)
	Commercial HMO	55 (15.7)
	Commercial high deductible	28 (8.0)
	Unknown	12 (3.4)
	Comprehensive*	7 (2.0)
	Medicare	0 (0.0)
Region n (%)	Northeast	72 (20.6)
	North Central	82 (23.4)
	South	117 (33.4)
	West	73 (20.9)
	Unknown	6 (1.7)
Underlying conditions		
Number of underlying conditions, mean (SD)		1.1 (0.7)
Number of underlying conditions, n (%)	1	188 (53.7)
	2	87 (24.9)
	3	8 (2.3)
	Other/unknown	67 (19.1)
Underlying conditions, n (%)†	Aplastic anemia	150 (42.9)
	Acute lymphoblastic leukemia	114 (32.6)
	Acute myeloid leukemia	69 (19.7)
	Other/unknown	67 (19.1)
	Myelodysplastic syndrome	27 (7.7)
	Chronic myeloid leukemia	10 (2.9)
	Hodgkin lymphoma	4 (1.1)
	Non-Hodgkin lymphoma	4 (1.1)
	Neuroblastoma	4 (1.1)
	Myelofibrosis	3 (0.9)
	Myeloma	1 (0.3)
	Ewing sarcoma	0 (0.0)

*Insurance plans without incentive for the patient to use a particular list of providers. Coverage is handled by only one policy, with a deductible and co-insurance.
†The percentages are not mutually exclusive, as patients can have more than one underlying condition.
Allo-HCT, allogeneic hematopoietic cell transplantation; HMO, Health Maintenance Organization; PPO/POS, Preferred Provider Organization/Point-of-Service; SD, standard deviation.

Rate of AdV infection

- 34 of 350 (9.7%) allo-HCT recipients had at least one ICD-9 code for AdV infection during the 365 day post-transplant period (**Figure 1**).
- The incidence rate of AdV infection was 0.13 infection per patient-year in the 365 days following transplant in the pediatric cohort who received an allo-HCT.
- The setting of first AdV infection diagnosis among the 34 patients with an ICD-9 code for AdV infection during the 365 day post-transplant period is reported in **Figure 2**. The first AdV infection diagnosis code was documented:
 - in 12 of 34 (35.3%) patients during the initial transplant admission
 - in 12 of 34 (35.3%) patients during a subsequent readmission
 - in 10 of 34 (29.4%) patients following a healthcare visit in another place of service (i.e., a non-hospital setting).

Figure 1. Incidence of diagnosed adenovirus infection in pediatric allogeneic hematopoietic cell transplantation recipients within the 365 day post-transplantation period

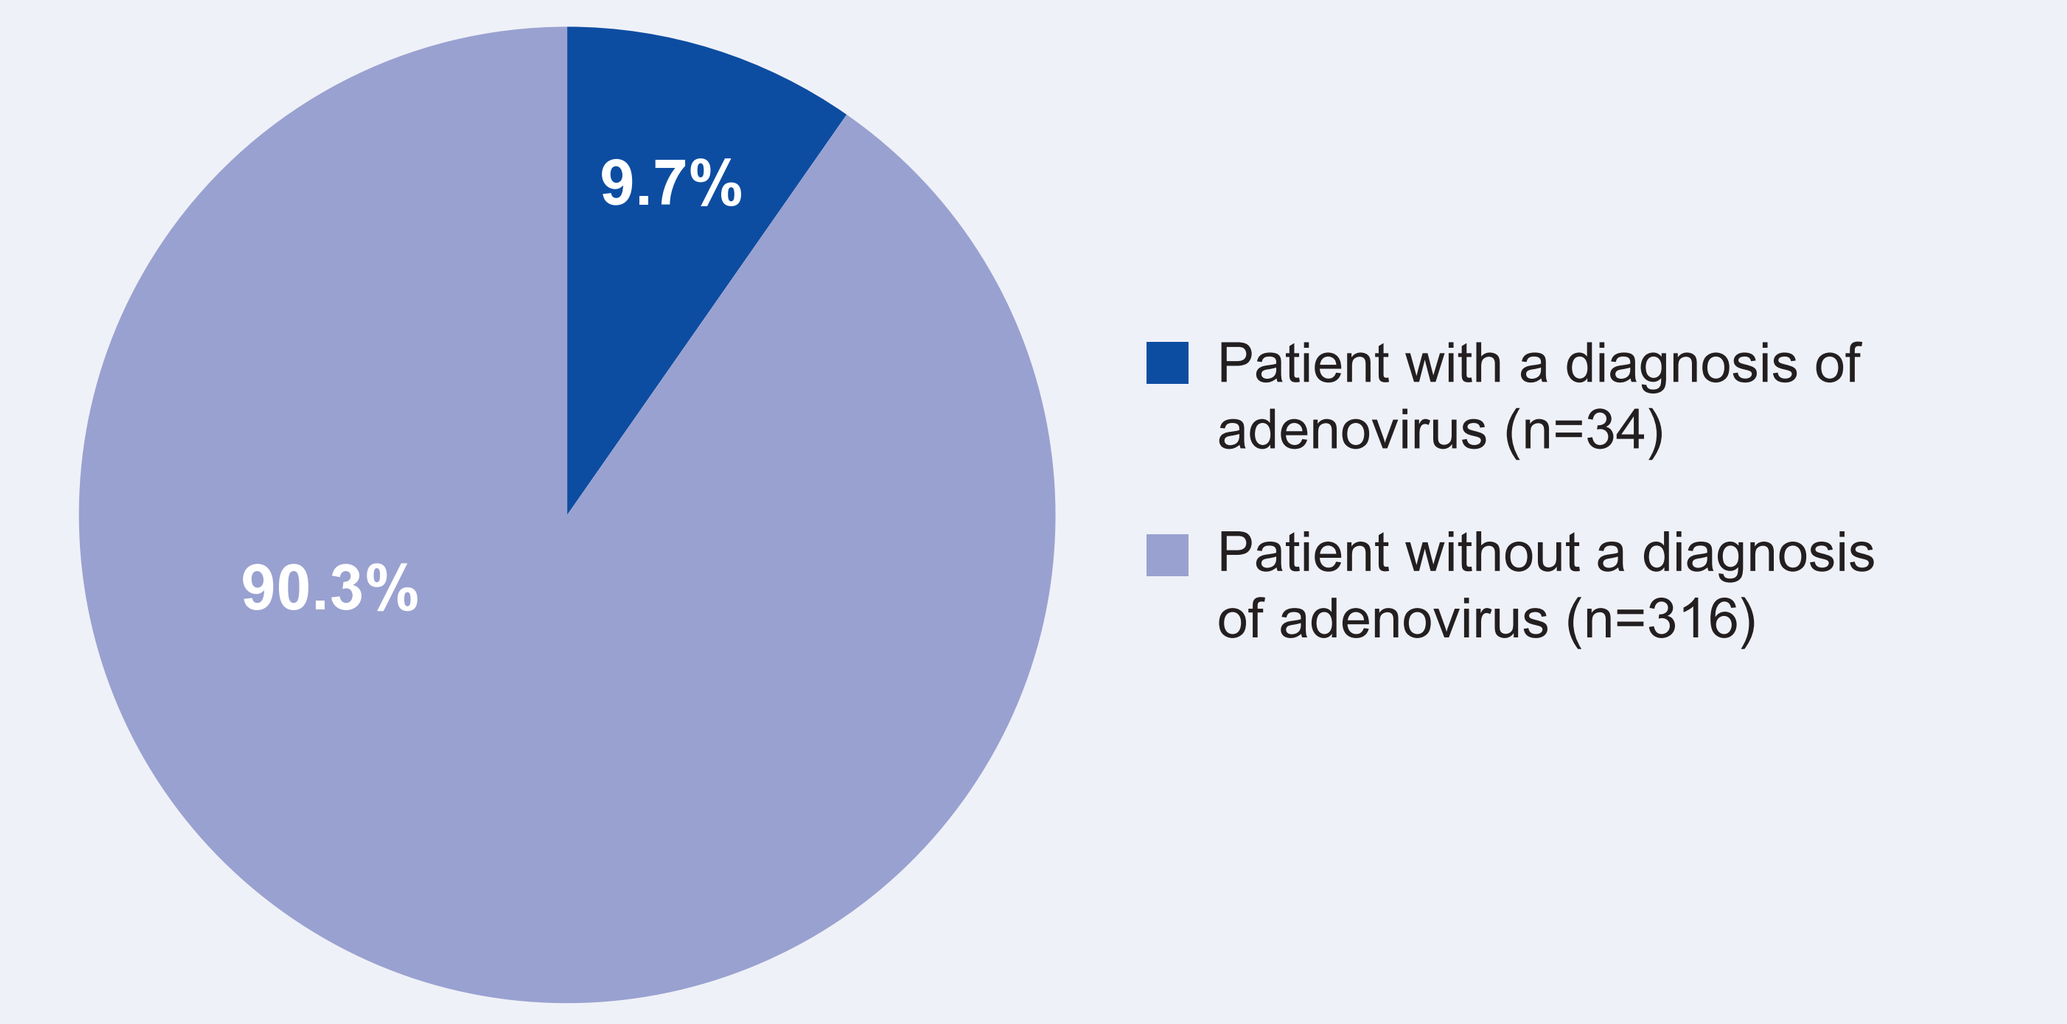
9.7%
90.3%
Patient with a diagnosis of adenovirus (n=34)
Patient without a diagnosis of adenovirus (n=316)

Figure 2. Documented setting of adenovirus infection diagnosis in pediatric allogeneic hematopoietic cell transplantation recipients

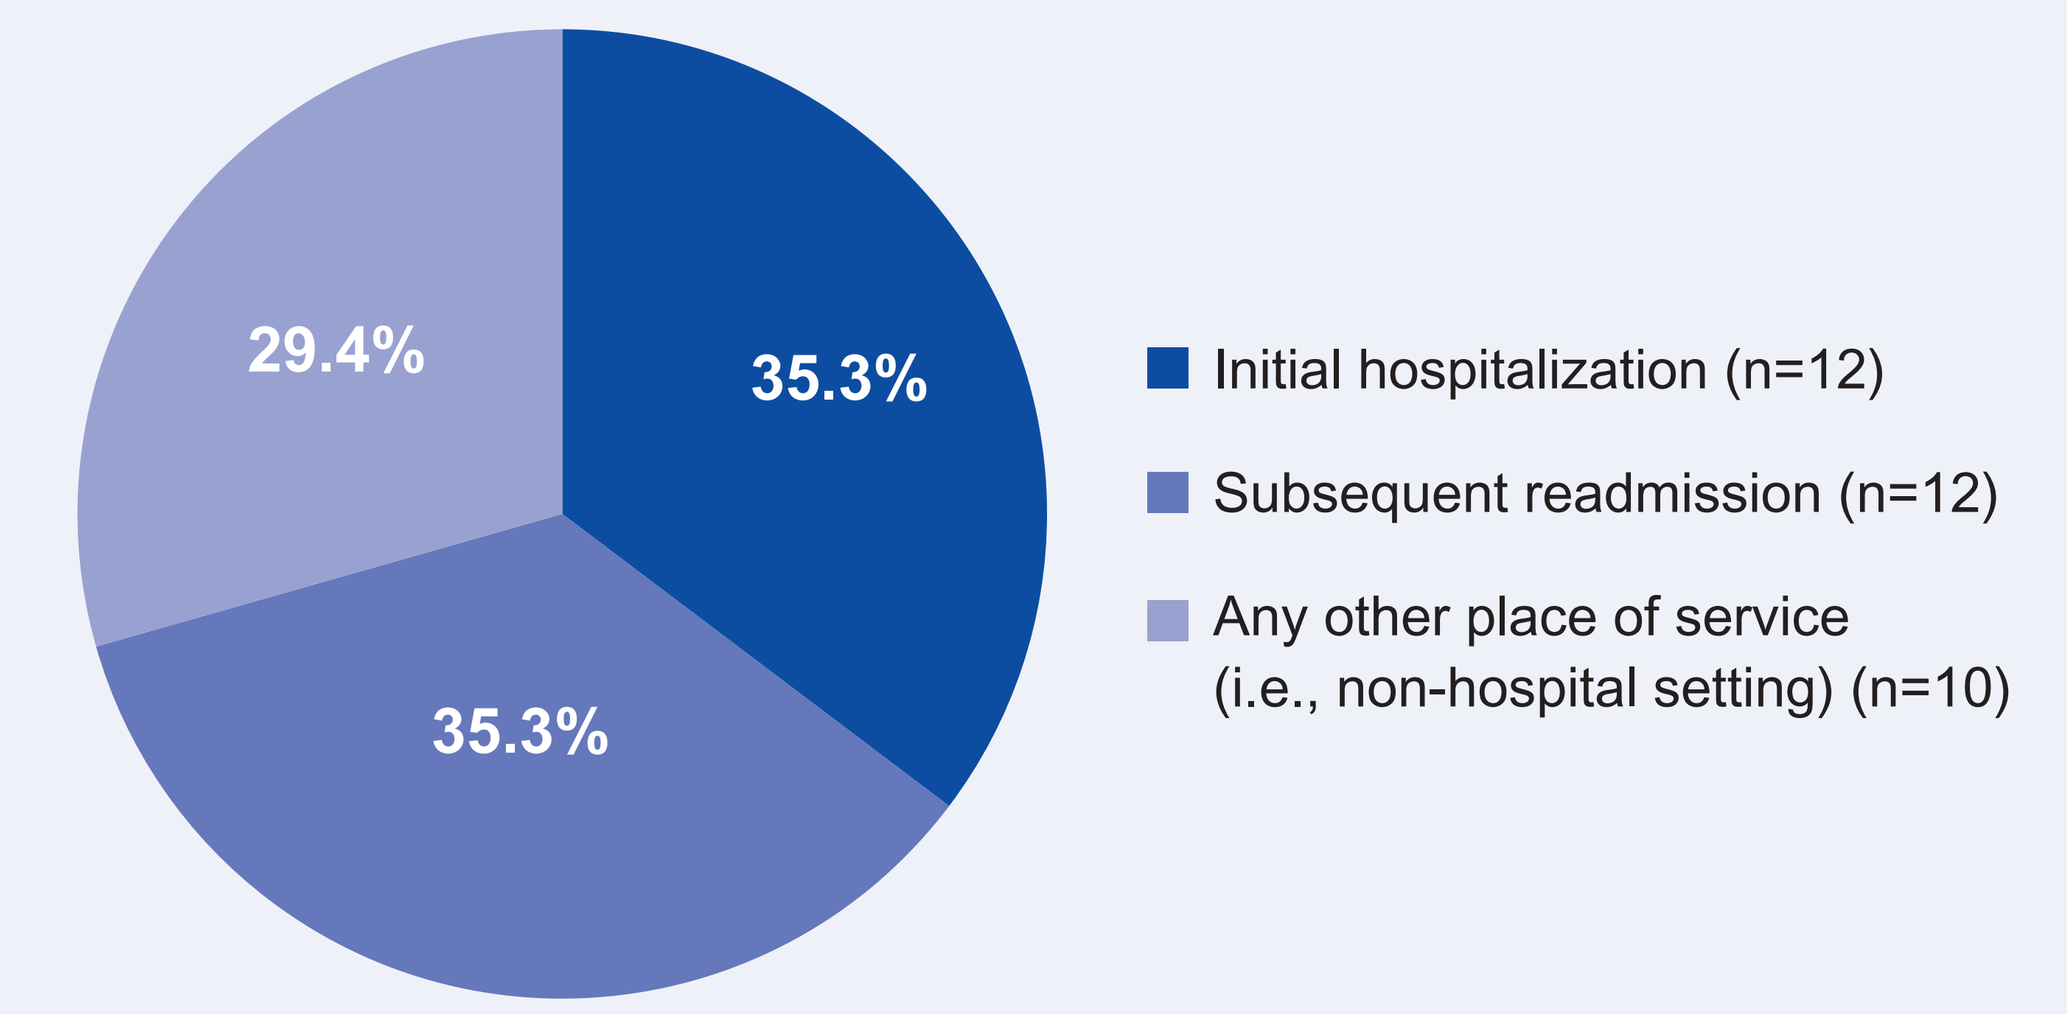
29.4%
35.3%
35.3%
Initial hospitalization (n=12)
Subsequent readmission (n=12)
Any other place of service (i.e., non-hospital setting) (n=10)

Figure 3. Distribution of time to first adenovirus infection diagnosis in pediatric allogeneic hematopoietic cell transplantation recipients

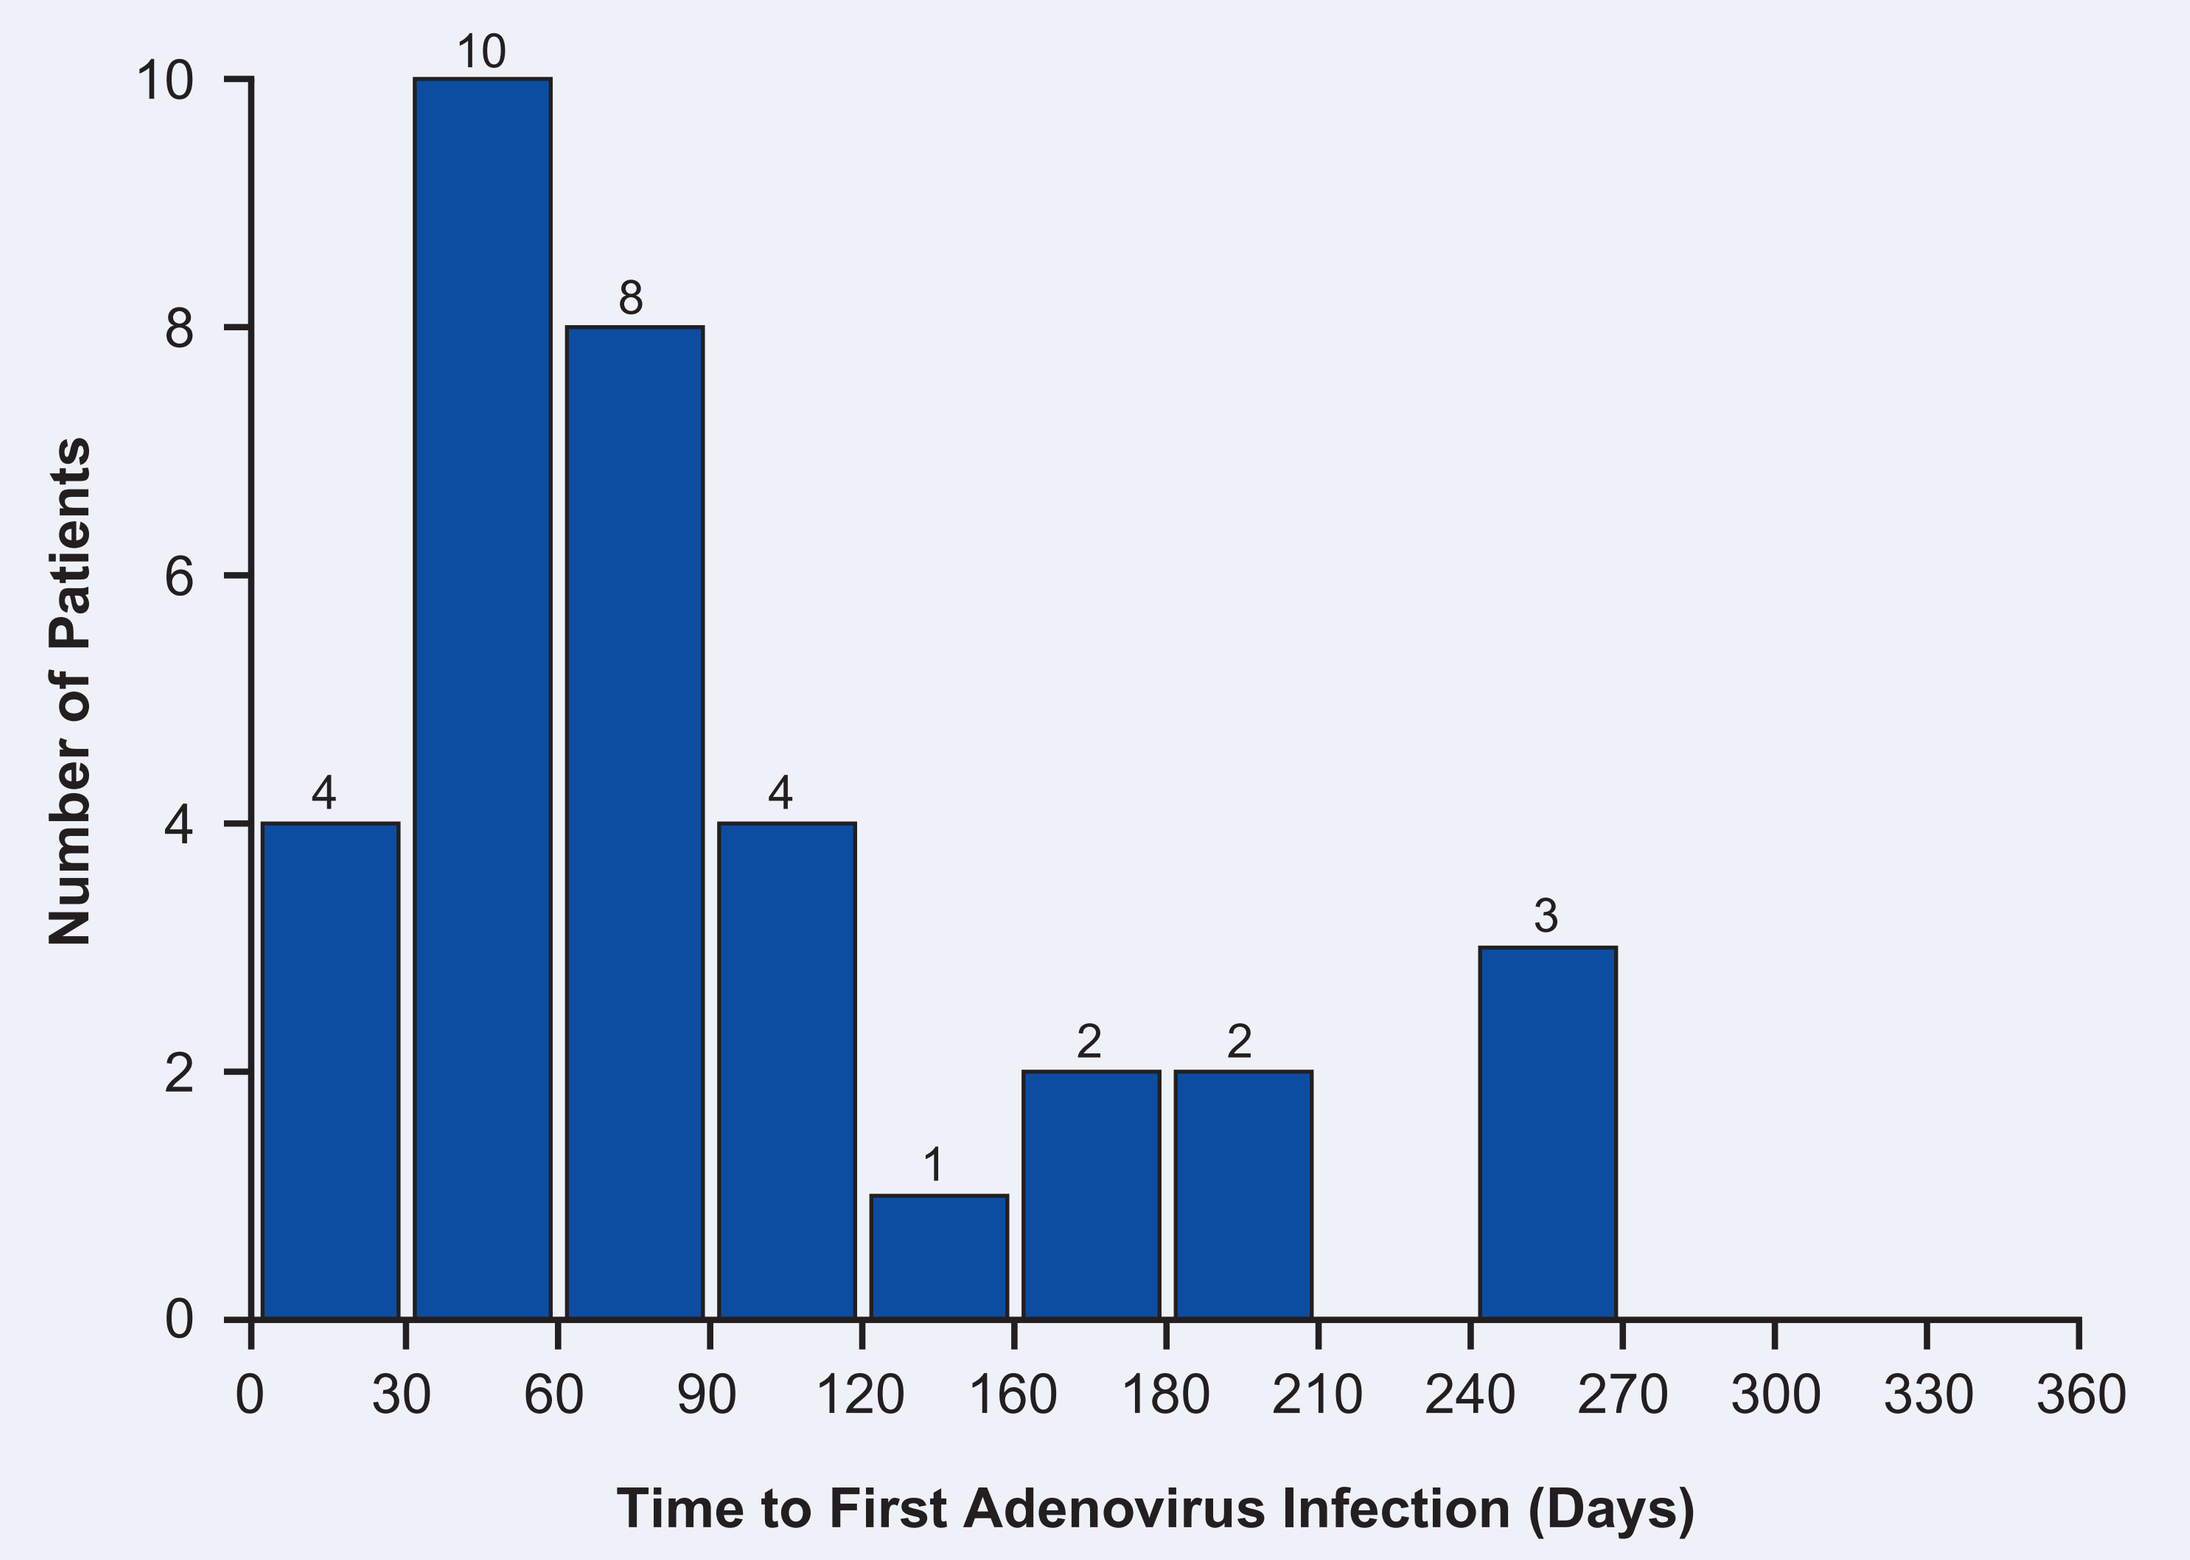
Number of Patients
0
2
4
6
8
10
4
10
8
4
1
2
2
3
0 30 60 90 120 160 180 210 240 270 300 330 360
Time to First Adenovirus Infection (Days)

- Among the 34 patients with an ICD-9 code for AdV infection, mean (SD) time to the first AdV infection was 91.2 (70.7) days and median time was 62 days (**Table 2, Figure 3**).

Table 2. Time to first adenovirus (AdV) infection diagnosis in pediatric allogeneic hematopoietic cell transplantation (allo-HCT) recipients

Time to first AdV infection diagnosis (days)	Pediatric allo-HCT recipients with at least one ICD-9 code for an AdV infection (n=34)
Mean (SD)	91.2 (70.7)
Median (Q1–Q3)	62 (43–119)

ICD-9, International Classification of Diseases, Ninth Revision; Q, quartile; SD, standard deviation.

LIMITATIONS

The true incidence of pediatric AdV infection may be higher as it is possible that not all AdV infections were detected. This could be due to a lack of specific screening for AdV, the potential use of non-specific ICD-9 codes in the insurance claims, and under-coding of identified infections.

CONCLUSIONS

- **Approximately 1 in 10 pediatric allo-HCT recipients from this US healthcare database were documented as having an ICD-9 diagnosis code for AdV infection within the first year post-allo-HCT.**
- **AdV infection tends to occur within the first few months after allo-HCT.**
- **Based on these findings, and given the high mortality rate reported for pediatric HCT recipients with disseminated AdV infection,[1] proactive screening should be considered, especially in the first few months after transplant.**

REFERENCE

1. George DE, et al. *Br J Haematol* 2012;156:99–108.

ACKNOWLEDGMENTS

Aminata Thiam, formerly of LASER Analytica, assisted with the development of the poster. Editorial assistance was provided by Lisa Jolly and Katrina Rimmer of Caudex, Oxford, UK, supported by Chimerix, Durham, NC.

DISCLOSURES

EM is an employee of Chimerix, Durham, NC. LS, Y-CL, VP, and RC are employees of LASER Analytica, New York, NY, which received research funds from Chimerix, Durham, NC, to conduct this database analysis, and for development of this poster.

BMT Tandem Meetings; February 18–22, 2016; Honolulu, Hawaii