First-Year Incidence of Double-Stranded DNA Virus Infections Following Hematopoietic Cell Transplantation

Essy Mozaffari,[1] PharmD, MPH, MBA; Yi-Chien Lee,[2] MS; Lee Stern,[2] MS; Vishal Patel,[2] BS; Roman Casciano,[2] MSc

[1]Chimerix, Durham, NC, USA; [2]LASER Analytica, New York, NY, USA

Presenting author contact details
E Mozaffari
Chimerix Inc.
2505 Meridian Parkway, Suite 340
Durham, NC 27713
Tel: (919) 806-1074
Fax: (919) 806-1146
email: emozaffari@chimerix.com

218

INTRODUCTION

- Immunosuppression during hematopoietic cell transplantation (HCT) increases the risk of opportunistic infections.
- Fewer viable options are available to prevent and treat double-stranded DNA (dsDNA) viral infections compared to other pathogens.
- This study describes the first-year incidence of dsDNA viral infections in US health insurance plan enrollees who received an allogeneic or autologous HCT.

METHODS

Data source

- Data for this study were extracted from the MarketScan Research Databases, which contain healthcare insurance claims records for commercial and Medicare patients.

Patients

- Patients were included in the study if they had a relevant International Classification of Diseases, Ninth Revision (ICD-9) or Current Procedural Terminology procedure code for an allogeneic or autologous HCT between June 26, 2010, and June 30, 2014.
- Patients must have had at least 360 days of health plan enrollment prior to the HCT procedure.
- No minimum health plan enrollment was required post-procedure.
- The first admission for an HCT procedure for each patient was defined as the index procedure.

Baseline characteristics

- Demographic and clinical characteristics reported in the MarketScan Research Databases were compiled for the study population, including:
 - age at time of transplant
 - sex
 - health insurance type
 - region
 - underlying conditions for which the transplant was performed.

dsDNA Viral Infection

- Patients with dsDNA viral infection were identified by having at least one ICD-9 diagnosis code for at least one of the following viral infections documented in the health insurance claims records within 365 days following transplant:
 - cytomegalovirus (CMV)
 - adenovirus
 - BK virus
 - Epstein–Barr virus
 - herpes simplex viruses
 - varicella zoster virus
 - human papillomavirus
 - other human herpesviruses.
- Among those patients with evidence of dsDNA viral infection, the setting of the infection diagnosis was documented as occurring:
 - In the initial hospital admission, when the first dsDNA viral infection diagnosis was associated with the initial transplant admission.
 - In a subsequent hospital admission, when the first dsDNA viral infection diagnosis was associated with a readmission after discharge from the initial transplant admission.
 - In any other place of service when the first dsDNA viral infection diagnosis was not associated with their initial transplant admissions nor a subsequent hospital admission (i.e., a non-hospital setting).
- Time to first dsDNA viral infection diagnosis (days) was reported for patients with evidence of dsDNA viral infection.

Statistical analysis

- Mean (standard deviation [SD]) or n (%) were reported for baseline characteristics.
- dsDNA viral infection:
 - Number and percentage of patients with at least one ICD-9 code for dsDNA viral infection.
 - Incidence rate for dsDNA viral infection defined as the number of infections divided by total patient-years of follow-up.
 - Follow-up time is defined as time from day of transplant up to 365 days or end of continuous health insurance plan enrollment (whichever was earlier).
 - Setting of dsDNA viral infection diagnosis:
 - number and percentage of patients in each setting.
 - Time to dsDNA viral infection diagnosis:
 - mean (SD), median, first quartile (Q1) and third quartile (Q3)
 - Kaplan–Meier (KM) curves for time to dsDNA viral infection diagnosis.

RESULTS

Study population

- 3035 allogeneic (mean age 47.3 years, 56.9% male) and 4793 autologous (mean age 53.4 years, 59.9% male) HCT recipients were identified within the MarketScan Research Databases as meeting the inclusion criteria (**Table 1**).
 - Most allogeneic (n=1973, 65.0%) and autologous (n=2800, 58.4%) HCT recipients were on a commercial preferred provider organization or point-of-service (PPO/POS)-type insurance plan.
 - The majority of allogeneic and autologous HCT recipients had one or two pre-specified underlying conditions, with the most commonly identified conditions being:
 - aplastic anemia (n=1386, 45.7%) and acute myeloid leukemia (n=1221, 40.2%) for allogeneic HCT recipients
 - myeloma (n=2543, 53.1%) and aplastic anemia (n=1863, 38.9%) for autologous HCT recipients (**Table 1**).

Table 1. Characteristics of study population

Characteristics		Allogeneic HCT recipients (n=3035)	Autologous HCT recipients (n=4793)
Age (years), mean (SD)		47.3 (17.9)	53.4 (15.2)
Male, n (%)		1727 (56.9)	2873 (59.9)
Insurance plan type, n (%)	Commercial PPO/POS	1973 (65.0)	2800 (58.4)
	Commercial HMO	353 (11.6)	587 (12.3)
	Commercial high deductible	238 (7.8)	324 (6.8)
	Medicare	223 (7.4)	725 (15.1)
	Unknown	175 (5.8)	238 (5.0)
	Comprehensive*	73 (2.4)	119 (2.5)
Regions, n (%)	Northeast	669 (22.0)	1064 (22.2)
	North Central	736 (24.3)	1177 (24.6)
	South	975 (32.1)	1613 (33.7)
	West	602 (19.8)	822 (17.2)
	Unknown	53 (1.8)	117 (2.4)
Underlying conditions			
Number of underlying conditions, mean (SD)		1.4 (0.8)	1.3 (0.7)
Number of underlying conditions, n (%)	1	1320 (43.5)	2381 (49.7)
	2	1176 (38.8)	1701 (35.5)
	3	194 (6.4)	70 (1.5)
	4	16 (0.5)	3 (0.1)
	5	1 (0.0)	0 (0.0)
	Other/unknown	328 (10.8)	638 (13.3)
Underlying conditions, n (%)†	Aplastic anemia	1386 (45.7)	1863 (38.9)
	Acute myeloid leukemia	1221 (40.2)	85 (1.8)
	Acute lymphoblastic leukemia	479 (15.8)	26 (0.5)
	Myelodysplastic syndrome	452 (14.9)	11 (0.2)
	Non-Hodgkin lymphoma	379 (12.5)	1012 (21.1)
	Other/unknown	328 (10.8)	638 (13.3)
	Chronic myeloid leukemia	140 (4.6)	3 (0.1)
	Myelofibrosis	107 (3.5)	5 (0.1)
	Myeloma	98 (3.2)	2543 (53.1)
	Hodgkin lymphoma	54 (1.8)	353 (7.4)
	Neuroblastoma	6 (0.2)	93 (1.9)

*Insurance plans without incentive for the patient to use a particular list of providers. Coverage is handled by only one policy, with a deductible and co-insurance.
†The percentages are not mutually exclusive, as patient can have more than one underlying condition.
HCT, hematopoietic cell transplantation; HMO, Health Maintenance Organization; PPO/POS: Preferred Provider Organization/Point-of-Service; SD, standard deviation.

Rate of dsDNA viral infection

- 924 of 3035 (30.4%) allogeneic HCT recipients and 351 of 4793 (7.3%) autologous HCT recipients had at least one dsDNA viral infection during the 365 day post-transplant period (**Table 2**).
- The incidence rate of dsDNA viral infections in the 365 days after transplant was 0.46 and 0.10 infection per patient-year for the allogeneic and autologous HCT recipients, respectively.
- The setting of the first dsDNA viral infection diagnosis among the 924 allogeneic HCT recipients and 351 autologous HCT recipients with a dsDNA viral infection during the 365 day post-transplant period is reported in **Figure 1**.

Table 2. Incidence of diagnosis of double-stranded DNA viral infection within 365 days post-transplantation in allogeneic and autologous hematopoietic cell transplantation (HCT) recipients

	Allogeneic HCT recipients (n=3035)	Autologous HCT recipients (n=4793)
Patients with event, n (%)	924 (30.4)	351 (7.3)
Incidence rate (per patient-year)	0.46	0.10

Figure 1. Setting of the first double-stranded DNA viral infection diagnosis within 365 days post-transplantation in allogeneic and autologous hematopoietic cell transplantation (HCT) recipients

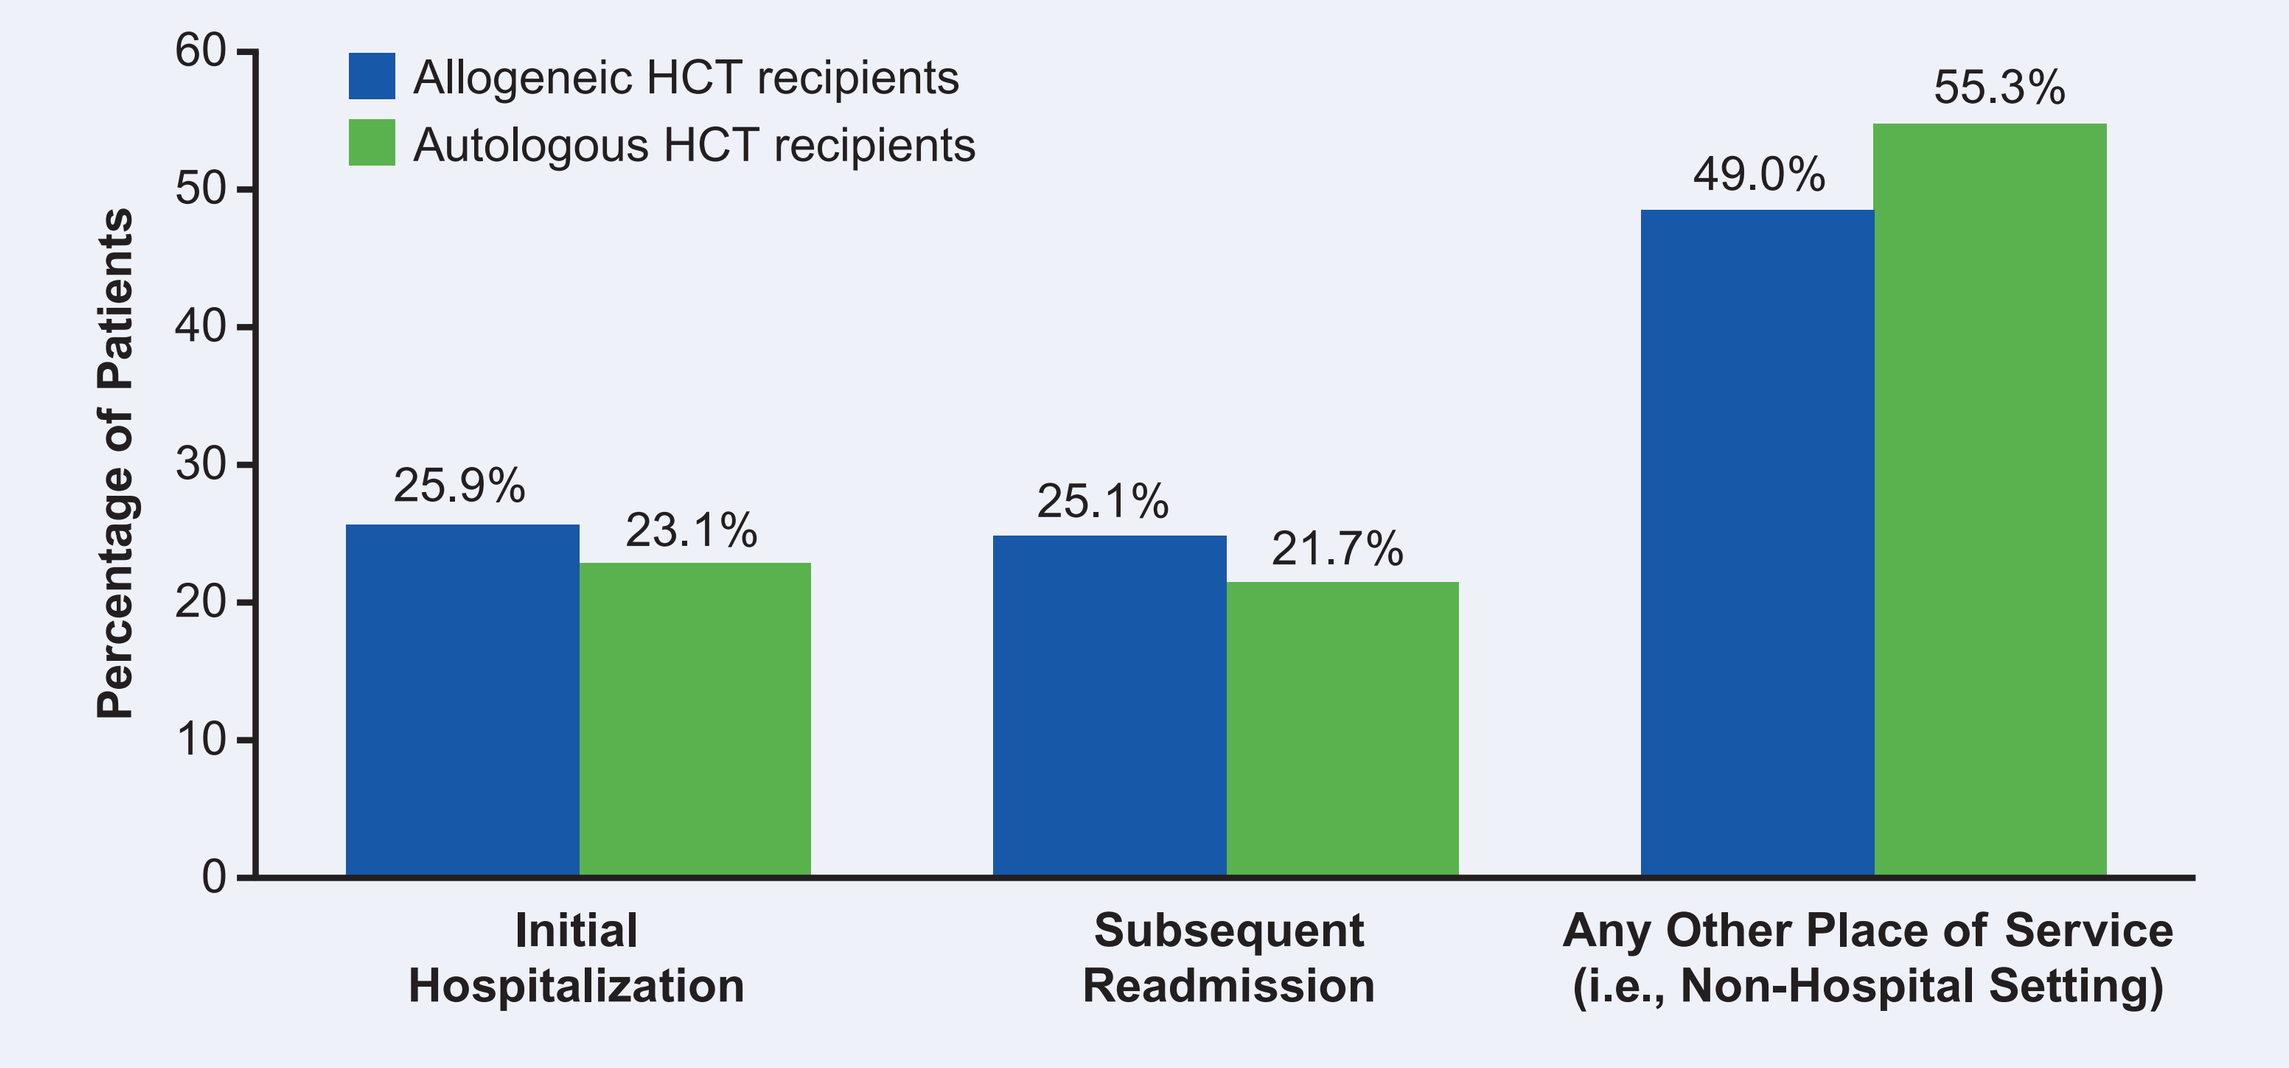

Percentage of Patients
60
50
40
30
20
10
0
Allogeneic HCT recipients
Autologous HCT recipients
25.9%
23.1%
25.1%
21.7%
49.0%
55.3%
Initial Hospitalization
Subsequent Readmission
Any Other Place of Service (i.e., Non-Hospital Setting)

- Among 924 allogeneic HCT recipients with dsDNA viral infections
 - 239 (25.9%) patients were diagnosed during the initial transplant admission
 - 232 (25.1%) patients were diagnosed during a subsequent readmission
 - 453 (49.0%) patients were diagnosed following a healthcare visit in another place of service (i.e., a non-hospital setting).
- Among 351 autologous HCT recipients with dsDNA viral infections
 - 81 (23.1%) patients were diagnosed during the initial transplant admission
 - 76 (21.7%) patients were diagnosed during a subsequent readmission
 - 194 (55.3%) patients were diagnosed following a healthcare visit in another place of service (i.e., a non-hospital setting).
- Among the 924 allogeneic and 351 autologous HCT recipients with an ICD-9 code for dsDNA viral infection, mean (SD) time to first dsDNA viral infection was 70.2 (73.1) and 112.1 (108.9) days, respectively. Median time to first infection was 50 and 75 days, respectively (**Table 3**). KM curves for time to dsDNA viral infections are shown in **Figure 2**.

Table 3. Time to first documented double-stranded DNA (dsDNA) viral infection diagnosis within 365 days post-transplantation in allogeneic and autologous hematopoietic cell transplantation (HCT) recipients

Time to first dsDNA viral infection diagnosis, days	Patient with at least one ICD-9 code for a dsDNA viral infection	
	Allogeneic HCT recipients (n=924)	Autologous HCT recipients (n=351)
Mean (SD)	70.2 (73.1)	112.1 (108.9)
Median (Q1–Q3)	50 (24–89)	75 (13–187)

ICD-9, International Classification of Diseases, Ninth Revision; Q, quartile; SD, standard deviation.

LIMITATIONS

The incidence of dsDNA viral infections based upon insurance claims database analysis may underestimate the true incidence of dsDNA viral infection, as there is a potential for underreporting due to lack of screening for specific pathogens, the use of non-specific ICD-9 diagnosis codes in insurance claims, and under-coding of identified infections.

Figure 2. Kaplan–Meier curves for time to dsDNA viral infection

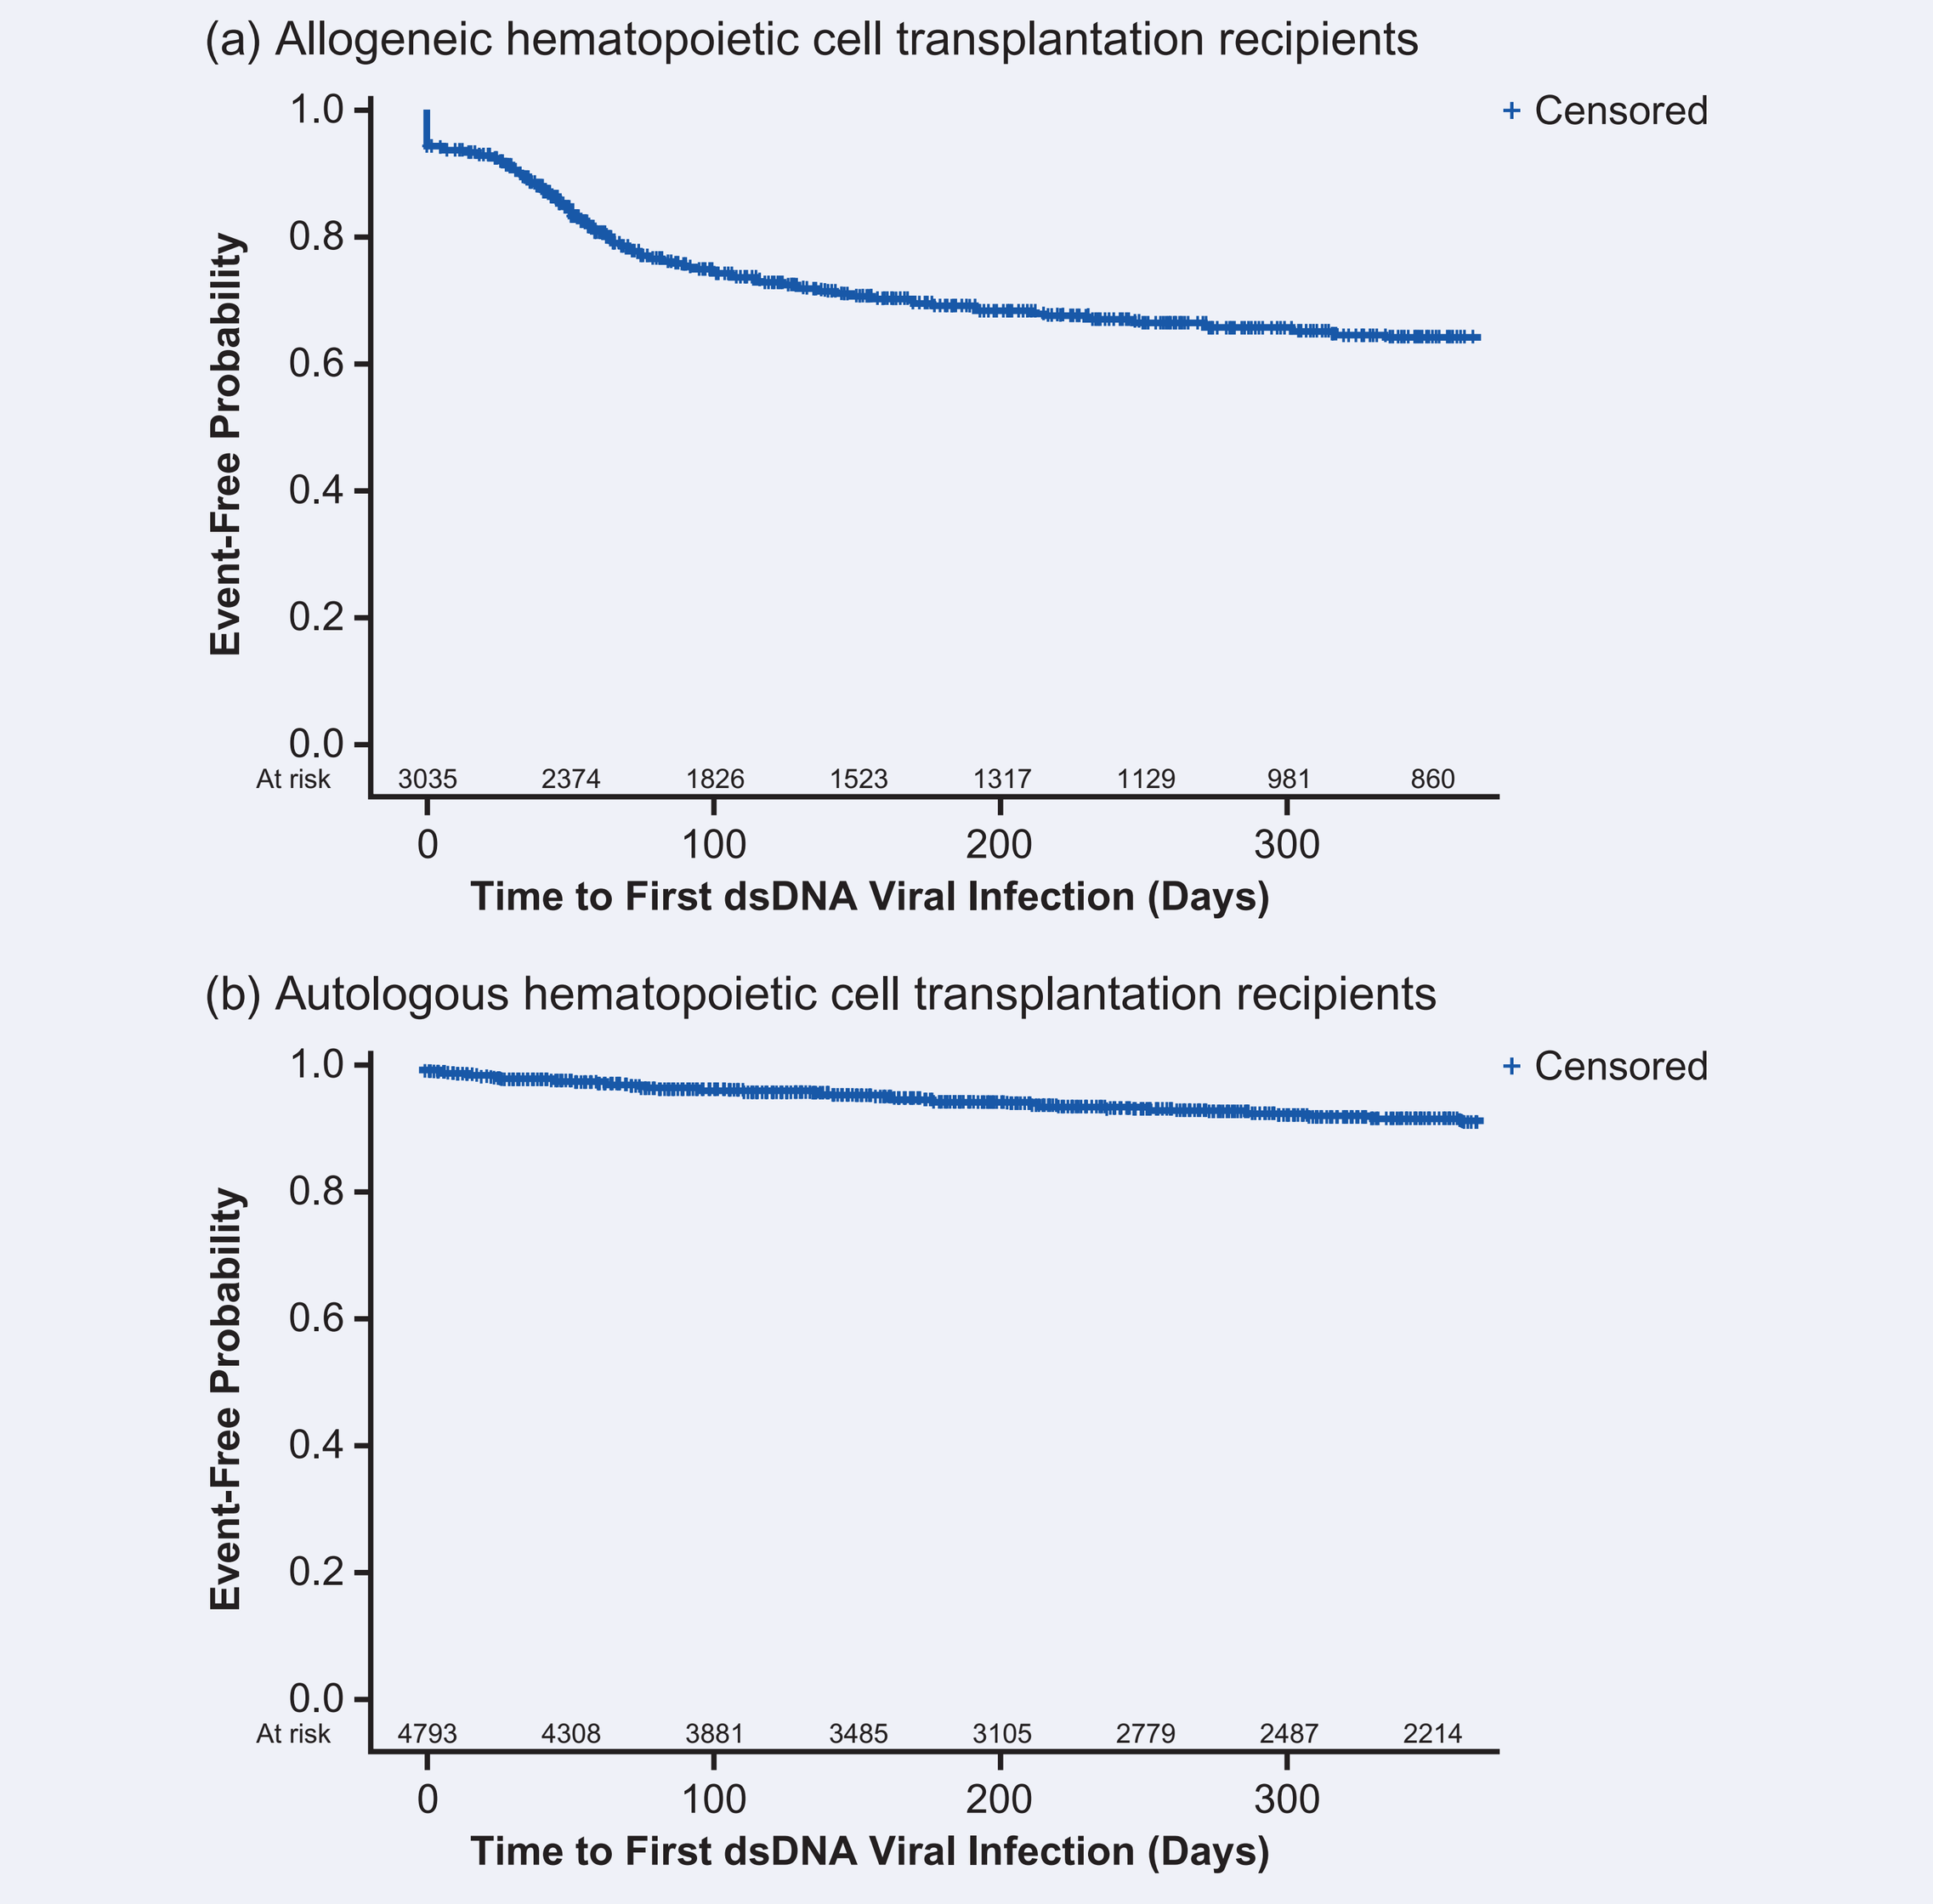

(a) Allogeneic hematopoietic cell transplantation recipients
+ Censored
Event-Free Probability
1.0
0.8
0.6
0.4
0.2
0.0
At risk
3035
2374
1826
1523
1317
1129
981
860
0
100
200
300
Time to First dsDNA Viral Infection (Days)
(b) Autologous hematopoietic cell transplantation recipients
+ Censored
Event-Free Probability
1.0
0.8
0.6
0.4
0.2
0.0
At risk
4793
4308
3881
3485
3105
2779
2487
2214
0
100
200
300
Time to First dsDNA Viral Infection (Days)

CONCLUSIONS

- **The potential dsDNA viral infection burden is particularly high for allogeneic HCT recipients, with 3 of 10 patients diagnosed with at least one dsDNA viral infection during the 365 days following HCT.**
- **dsDNA viral infections among allogeneic HCT recipients tend to occur within the first few months post HCT.**
- **Almost half of the dsDNA viral infections were diagnosed during initial transplant admission or subsequent readmission.**

ACKNOWLEDGMENTS

Aminata Thiam, formerly of LASER Analytica, assisted with the development of the poster. Editorial assistance was provided by Lisa Jolly and Katrina Rimmer of Caudex, UK, supported by Chimerix, Durham, NC.

DISCLOSURES

EM is an employee of Chimerix, Durham, NC. Y-CL, LS, VP, and RC are employees of LASER Analytica, New York, NY, which received research funds from Chimerix, Durham, NC, to conduct this database analysis, and for development of this poster.

BMT Tandem Meetings; February 18–22, 2016; Honolulu, Hawaii