203

Clinical and Economic Consequences Associated with Cytomegalovirus Infection among Allogeneic Hematopoietic Cell Transplant Patients

Essy Mozaffari,[1] PharmD, MPH, MBA; Roman Casciano,[2] MSc; Lee Stern,[2] MS; Yi-Chien Lee,[2] MS; Vishal Patel,[2] BS

[1]Chimerix, Durham, NC; [2]LASER Analytica, New York, NY

Presenting author contact details
E Mozaffari
Chimerix Inc.
2505 Meridian Parkway, Suite 340
Durham, NC 27713
Tel: (919) 806-1074
Fax: (919) 806-1146
email: emozaffari@chimerix.com

INTRODUCTION

- Hematopoietic cell transplantation (HCT) is a potentially curative therapy for patients with certain high-risk hematologic diseases.[1]
- HCT recipients are at risk of severe morbidity and mortality from cytomegalovirus (CMV) infection due to the immunosuppression associated with the transplant.[2]
- In this study we assessed the health and healthcare consequences among allogeneic HCT (allo-HCT) patients being actively managed for CMV viremia or infection.

METHODS

Data source

- Data for this study were extracted from the MarketScan Research Databases, which contain electronic healthcare insurance claims records for commercial and Medicare patients.

Patients

- Patients were included in the study if they had a relevant International Classification of Diseases, Ninth Revision (ICD-9) or Current Procedural Terminology procedure code for an allo-HCT between June 26, 2010, and June 30, 2014.
- Patients must have had at least 360 days of health insurance plan enrollment prior to the allo-HCT procedure.
- No minimum health plan enrollment was required post-procedure.
- The first admission for an HCT procedure for each patient was defined as the index procedure.

CMV Population

- Patients were assigned to the CMV group for the base case analyses if they met one of the following two criteria post-allo-HCT through 365 days or to the end of their follow-up, whichever was earlier:
 - received treatment for CMV (valganciclovir, ganciclovir, foscarnet, cidofovir, CMV intravenous immune globulin) within 30 days of CMV infection diagnosis
 - had a hospitalization with a discharge diagnosis for CMV.

Non-CMV Population

- Patients were assigned to the non-CMV group for the base case analyses if they met one of the following two criteria post-allo-HCT through 365 days or to the end of their follow-up, whichever was earlier:
 - No documented evidence of CMV or other double-stranded DNA (dsDNA) viruses or CMV treatments.
 - Other types of dsDNA viral infections include the following: adenovirus, BK virus, Epstein–Barr virus, herpes simplex viruses, varicella zoster virus, human papillomavirus, and other human herpesviruses.
- Had only an outpatient diagnosis code (i.e., not associated with any hospitalization) for CMV or other dsDNA viruses in the absence of any evidence for CMV treatments.

Exclusion Criteria (for Base Case Analyses)

- A number of patients' retrospective insurance claims histories were difficult to discern, making appropriate assignment to the CMV or non-CMV groups uncertain. To this end, these patients were excluded from the base case analyses and the impact of this exclusion was tested in a sensitivity analysis.
 - Excluded group 1 (treatment only):
 - Patients with CMV treatments in their insurance claims records, but without evidence of either a CMV-related hospitalization or an outpatient diagnosis of CMV occurring within 30 days of the claims for CMV treatment.
 - Excluded group 2 (other dsDNA viral infections):
 - Patients with evidence of other types of dsDNA viral infections but with no diagnosis codes for CMV.
 - Patients who received treatment for CMV within 30 days of infection diagnosis for other types of dsDNA viral infections or had a hospitalization with any diagnosis for other types of dsDNA viral infections.

Patient Demographics

- The following characteristics were extracted for the CMV and non-CMV groups:
 - age at time of transplant and percentage of pediatric patients (defined as age <20 years old)
 - sex
 - region
 - health insurance type
 - underlying conditions for which the transplant was performed
 - total healthcare reimbursements in the year prior to the transplant procedure.

Outcome measures

- The following outcomes during the follow-up period were measured for the CMV and non-CMV groups. Follow-up period is defined as time from day of transplant up to 365 days or the end of continuous health insurance plan enrollment (whichever was earlier).
 - Total hospitalization days:
 - total number of days patients were hospitalized during the follow-up period.
 - Mortality:
 - any hospital discharge designation of "death" or "hospice" (with transfer to hospice presumed to be a proxy for mortality).
 - Total healthcare reimbursements:
 - all reimbursed amounts as reported in the insurance claims during the follow-up period
 - reimbursed amounts are presented in 2014 US dollars, adjusted using the medical care component of the US Consumer Price Index.[3]

Statistical analysis

- Mean (standard deviation [SD]) or n (%) were reported for baseline characteristics.
- Mean (SD) and median (quartile [Q]1–Q3) were reported for total hospitalization days and total healthcare reimbursements.
 - Variation in follow-up time was accounted for by annualizing total hospitalization days and total healthcare reimbursements for each patient.
 - Annualization of hospitalization days was calculated by dividing total costs or hospitalization days by follow-up duration.
 - Annualization of healthcare reimbursement was calculated by adding transplant day reimbursements to the quotient of post-transplant reimbursements divided by follow-up duration.
 - Generalized linear models with negative binomial (for total hospitalization days) and gamma distribution (for total reimbursements) were used to compare CMV and non-CMV groups.
- For mortality, the number and percentage of patients who died or were transferred to a hospice were quantified. Mortality rate was reported and Cox proportional hazards regression was used to compare the mortality rate between CMV and non-CMV groups.

Sensitivity analysis

- A sensitivity analysis was performed to test the impact of the exclusion criteria applied in the base case analyses.
- Three different scenarios were assessed as part of the sensitivity analysis:
 - Scenario 1: including excluded group 1 in the CMV group
 - Scenario 2: including excluded group 1 in the non-CMV group
 - Scenario 3: including excluded group 1 in the CMV group and including excluded group 2 in the non-CMV group
 - This scenario includes all allo-HCT recipients identified in the database without excluding any patients from the analysis.
- Each of the analyses undertaken for the base case analyses were performed for the three scenarios outlined above to evaluate if they had an impact on the comparison between CMV and non-CMV groups.

RESULTS

Patient population

- **Table 1** details the baseline characteristics of the base case population, which consisted of 546 patients with CMV and 2096 without CMV:
 - The mean age was 47.7 years for both CMV and non-CMV groups.
 - 292 of 546 (53.5%) patients in the CMV group and 1209 of 2096 (57.7%) patients in the non-CMV group were male.
 - The majority of patients were on a commercial Preferred Provider Organization/Point-of-Service (PPO/POS)-type health plan in both the CMV (65.4%) and non-CMV groups (64.9%).
 - Mean (median) total healthcare reimbursements during the 1-year period prior to the allo-HCT procedure were $281,037 ($225,055) for the CMV group and $237,348 ($194,705) for the non-CMV group.
 - In both CMV and non-CMV groups, most patients had one or two pre-specified underlying conditions.
 - The most commonly identified conditions were aplastic anemia (277 of 546 [50.7%] patients in the CMV group; 913 of 2096 [43.6%] patients in the non-CMV group) and acute myeloid leukemia (107 of 546 [19.6%] patients in the CMV group; 305 of 2096 [14.6%] patients in the non-CMV group).

Table 1. Patient characteristics for the base case population

	CMV (n=546)	Non-CMV (n=2096)
Age (years), mean (SD)	47.7 (18.1)	47.7 (17.5)
Proportion of pediatric patients (<20 years), n (%)	73 (13.4)	216 (10.3)
Male sex, n (%)	292 (53.5)	1209 (57.7)
Region, n (%)		
Northeast	94 (17.2)	484 (23.1)
North Central	114 (20.9)	538 (25.7)
South	212 (38.8)	631 (30.1)
West	116 (21.3)	407 (19.4)
Unknown	10 (1.8)	36 (1.7)
Total reimbursement in the 365 days prior to admission for allo-HCT procedure, mean (SD)	$281,037 (237,821)	$237,348 (199,681)
Number of underlying conditions, n (%)		
1	251 (46.0)	915 (43.7)
2	218 (39.9)	787 (37.6)
3	44 (8.1)	116 (5.5)
4	6 (1.1)	9 (0.4)
5	0 (0.0)	1 (0.1)
Type of underlying condition, n (%)		
Aplastic anemia	277 (50.7)	913 (43.6)
Acute myeloid leukemia	234 (42.9)	811 (38.7)
Acute lymphoblastic leukemia	107 (19.6)	305 (14.6)
Myelodysplastic syndrome	89 (16.3)	316 (15.1)
Non-Hodgkin lymphoma	72 (13.2)	250 (11.9)
Other/unknown	27 (5.0)	268 (12.8)
Chronic myeloid leukemia	23 (4.2)	96 (4.6)
Myelofibrosis	20 (3.7)	73 (3.5)
Hodgkin lymphoma	7 (1.3)	39 (1.9)
Neuroblastoma	1 (0.2)	3 (0.1)
Insurance plan type, n (%)		
Commercial PPO/POS	357 (65.4)	1361 (64.9)
Commercial HMO	63 (11.5)	231 (11.0)
Medicare	53 (9.7)	152 (7.3)
Commercial high deductible	32 (5.9)	175 (8.4)
Unknown	32 (5.9)	119 (5.7)
Comprehensive	9 (1.7)	58 (2.8)

Allo-HCT, allogeneic hematopoietic cell transplantation; CMV, cytomegalovirus; HMO, health maintenance organization; PPO/POS, Preferred Provider Organization/Point-of-Service; SD, standard deviation.

Base case analyses

- Tables 2–4 summarize the results of the base case analyses for the CMV and non-CMV groups.

Total Hospitalization Days Per Patient-Year During the 1-Year Post-Transplant Period

- The mean (median) number of total hospitalization days per patient-year was significantly higher among the CMV group (95.3 [67] days per patient-year) in comparison to the non-CMV group (78.5 [45] days per patient-year, p<0.0001, **Table 2**).

Table 2. Total hospitalization days per patient-year in patients with and without cytomegalovirus (CMV) infection during the 1-year period following allogeneic hematopoietic cell transplantation

		CMV (n=546)	Non-CMV (n=2096)	p value
Observed total hospitalization days	**Mean (SD)**	61.6 (44.3)	36.5 (31.9)	NA*
	Median (Q1–Q3)	50 (32–76)	30 (19–49)	
Annualized total hospitalization days	**Mean (SD)**	95.3 (83.0)	78.5 (92.5)	<0.0001
	Median (Q1–Q3)	67 (41–119)	45 (24–93)	

*Statistical comparison was only performed for total hospitalization days per patient-year to account for varying follow-up times.
NA, not available; Q, quartile; SD, standard deviation.

Mortality Rate Per Patient-Year During the 1-Year Post-Transplant Period

- Similarly, the mean rate of deaths was significantly higher in the CMV group (0.36 death per patient-year) in comparison to the non-CMV group (0.23 death per patient-year, p<0.0001, **Table 3**).

Total Healthcare Reimbursements During the 1-Year Post-Transplant Period

- Healthcare reimbursements were also higher in the CMV group, with mean (median) total healthcare reimbursements per patient-year during the 1-year post transplant of $712,164 ($512,441) in the CMV group compared to $427,085 ($306,100) in the non-CMV group (p<0.0001, **Table 4**).

Table 3. Mortality rate per patient-year in patients with and without cytomegalovirus (CMV) infection during the 1-year period following allogeneic hematopoietic cell transplantation

	CMV (n=546)	Non-CMV (n=2096)	p value
Patients who died or transferred to hospice, n (%)	130 (23.8)	298 (14.2)	NA*
Rate per patient-year	0.36	0.23	<0.0001

*Statistical comparison was only performed for mortality rate per patient-year to account for varying follow-up times.
NA, not available.

Table 4. Total healthcare reimbursements in patients with and without cytomegalovirus (CMV) infection during the 1-year period following allogeneic hematopoietic cell transplantation

		CMV (n=546)	Non-CMV (n=2096)	p value
Observed total reimbursements, $	**Mean (SD)**	569,369 (492,903)	317,742 (281,893)	NA*
	Median (Q1–Q3)	444,677 (279,189–681,297)	258,963 (146,479–404,424)	
Annualized total reimbursements, $	**Mean (SD)**	712,164 (791,762)	427,085 (536,561)	<0.0001
	Median (Q1–Q3)	512,441 (325,756–864,792)	306,100 (186,665–503,182)	

*Statistical comparison was only performed for annualized total reimbursements to account for varying follow-up times.
NA, not available.

Sensitivity analyses

- The results of the four sensitivity analysis scenarios are provided in **Figures 1–3**. Overall sensitivity analyses were consistent with the results of the base case analyses.

*Total Hospitalization Days Per Patient-Year During the 1-Year Post-Transplant Period (**Figure 1**)*

- Total hospitalization per patient-year during the 1-year post-transplant period ranged from 86.5 to 95.3 days in the CMV group and from 77.1 to 81.0 days in the non-CMV group.
- Total hospitalization days per patient-year were significantly higher in the CMV group versus the non-CMV group in all four scenarios, consistent with the base case results.

Figure 1: Hospitalization days in patients with and without cytomegalovirus (CMV) infection during the 1-year period following allogeneic hematopoietic cell transplantation

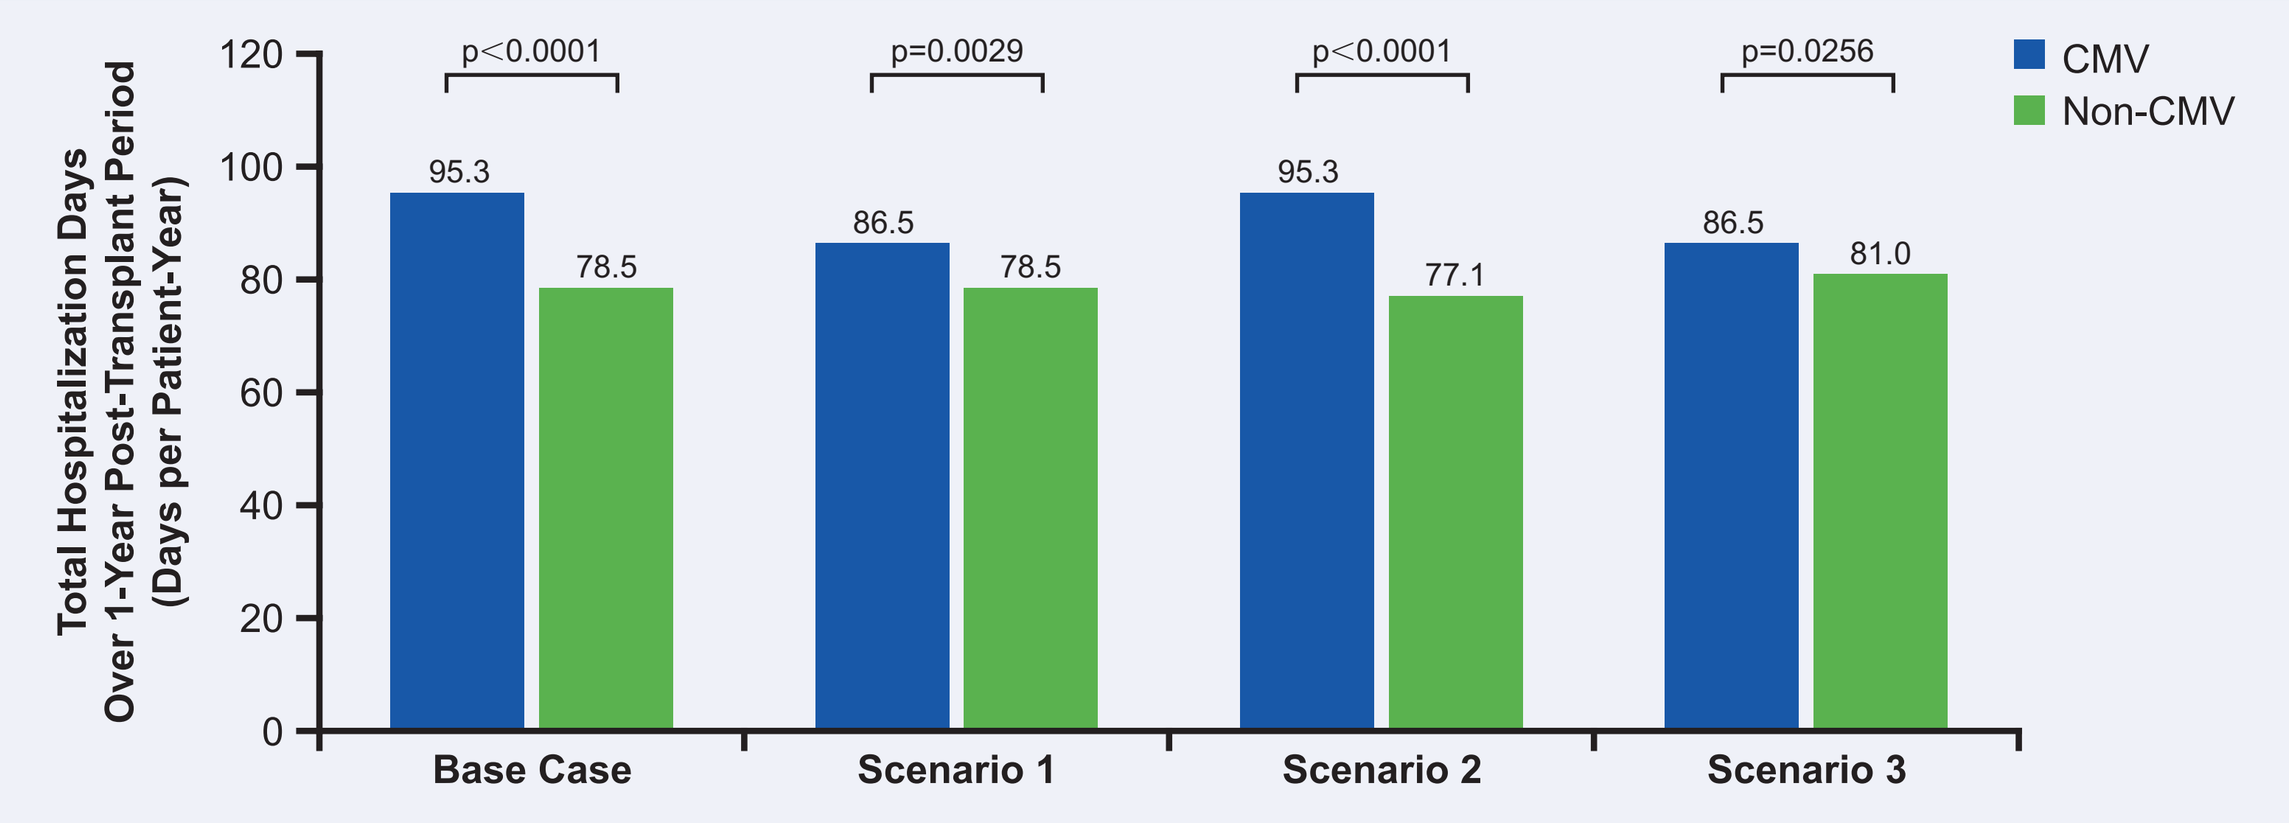

Total Hospitalization Days Over 1-Year Post-Transplant Period (Days per Patient-Year)
120
100
80
60
40
20
0
p<0.0001
p=0.0029
p<0.0001
p=0.0256
95.3
78.5
86.5
78.5
95.3
77.1
86.5
81.0
Base Case
Scenario 1
Scenario 2
Scenario 3
CMV
Non-CMV

*Mortality Rate Per Patient-Year During the1-Year Post-Transplant Period (**Figure 2**)*

- The rate of death per patient during the 1-year post-transplant period was between 0.3 to 0.4 in the CMV group and was 0.2 in all of the scenarios for the non-CMV group.
- As per the base case analyses, the rate of death was significantly higher in the CMV group versus the non-CMV group in all four scenarios.

Figure 2: Mortality rate per patient-year in patients with and without cytomegalovirus (CMV) infection during the 1-year period following allogeneic hematopoietic cell transplantation

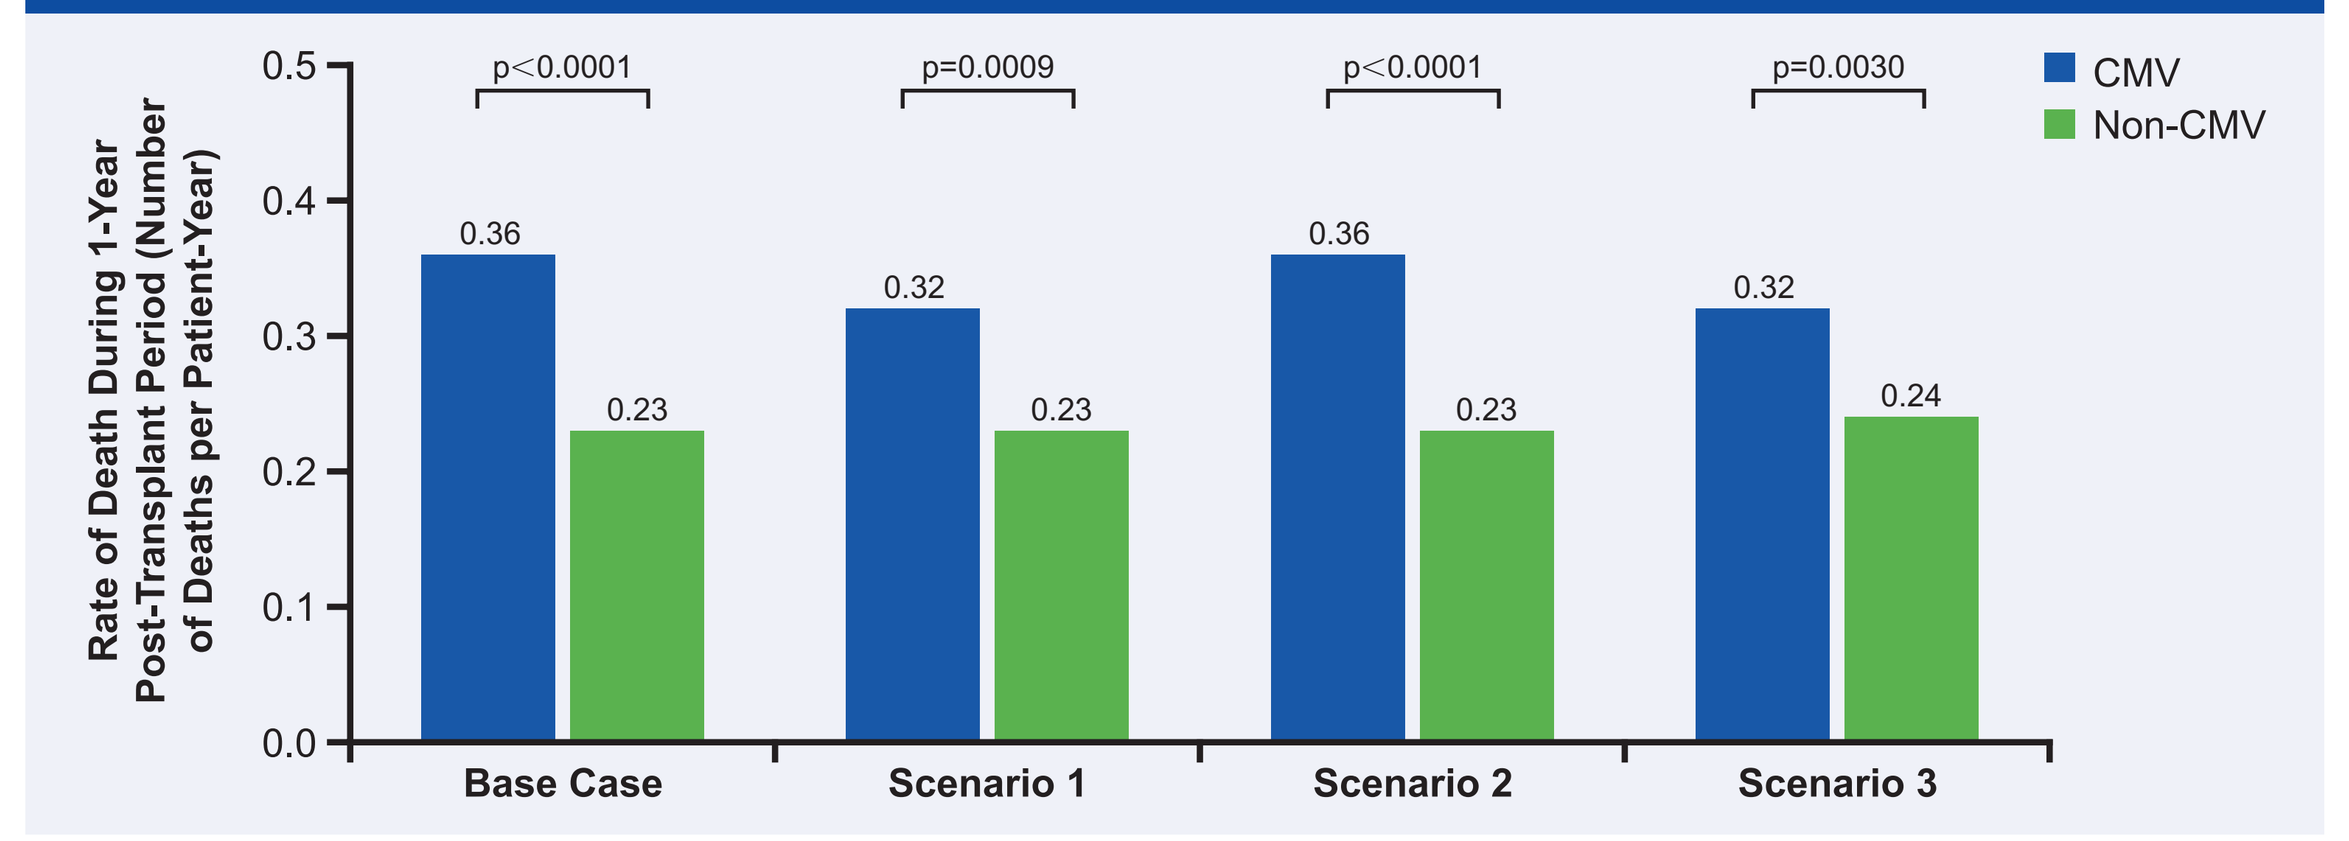

Rate of Death During 1-Year Post-Transplant Period (Number of Deaths per Patient-Year)
0.5
0.4
0.3
0.2
0.1
0.0
p<0.0001
p=0.0009
p<0.0001
p=0.0030
0.36
0.23
0.32
0.23
0.36
0.23
0.32
0.24
Base Case
Scenario 1
Scenario 2
Scenario 3
CMV
Non-CMV

*Total Healthcare Reimbursements Per Patient-Year During the 1-Year Post-Transplant Period (**Figure 3**)*

- Healthcare reimbursements per patient-year during the 1-year post-transplant period ranged between $639,466 and $712,164 for the CMV group and between $427,085 and $456,648 for the non-CMV group.
- Across all four scenarios, total healthcare reimbursements were significantly higher among patients in the CMV group versus patients in the non-CMV group.

Figure 3: Total healthcare reimbursements in patients with and without cytomegalovirus (CMV) infection during the 1-year period following allogeneic hematopoietic cell transplantation

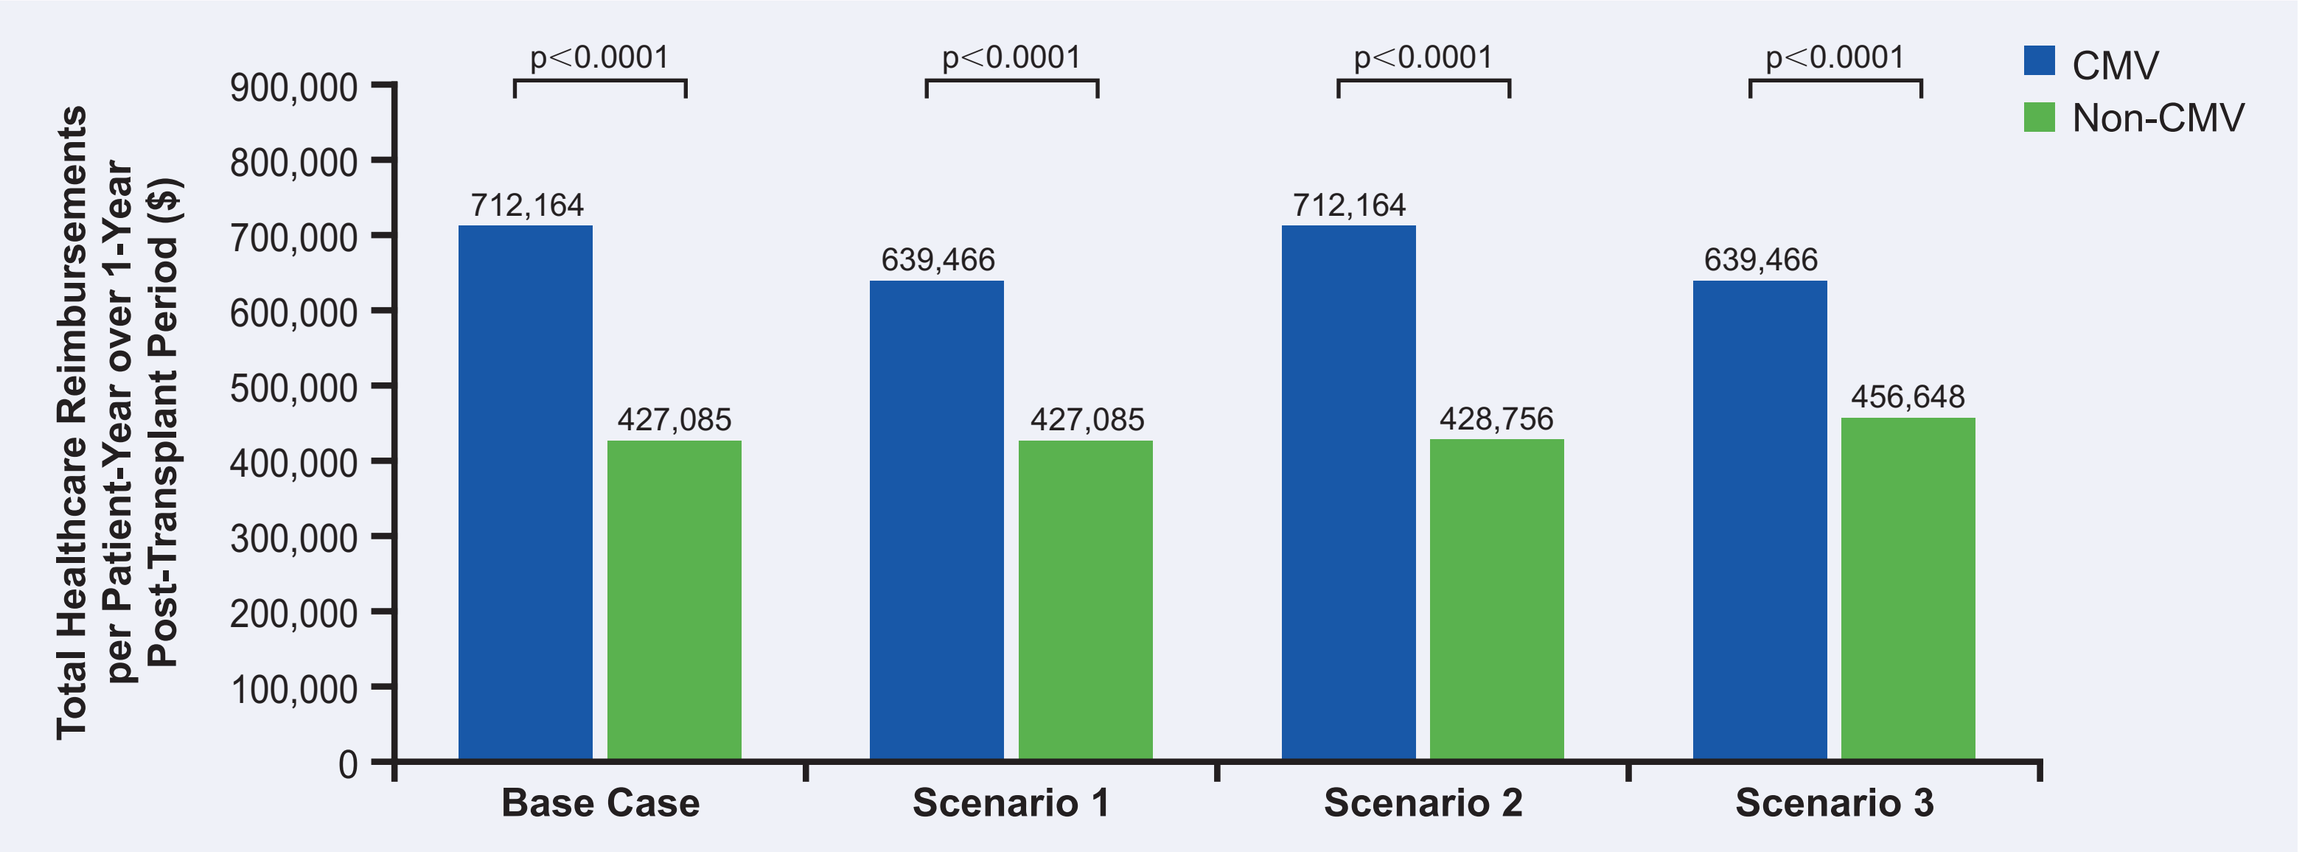

Total Healthcare Reimbursements per Patient-Year over 1-Year Post-Transplant Period ($)
900,000
800,000
700,000
600,000
500,000
400,000
300,000
200,000
100,000
0
p<0.0001
p<0.0001
p<0.0001
p<0.0001
712,164
427,085
639,466
427,085
712,164
428,756
639,466
456,648
Base Case
Scenario 1
Scenario 2
Scenario 3
CMV
Non-CMV

LIMITATIONS

A limitation of our study is potential under-coding and underreporting in the insurance claims records due to patient complexity and non-specific coding practices, which may underestimate the true incidence of CMV infection.

CONCLUSIONS

- **Our study demonstrated that patients who experience a CMV infection within 1 year following an allo-HCT procedure had significantly more hospitalization days and higher mortality rates than those without CMV infection.**
- **Significantly higher healthcare reimbursement costs were also observed for patients with CMV in comparison to those without CMV.**
- **The results remained consistent when the analysis decision rules for group assignment were tested via scenario analyses.**

REFERENCES

1. Tyndall A, et al. *Bone Marrow Transplant* 1999;24:729–34.
2. Biron KK. *Antiviral Res* 2006;71:154–63.
3. United States Department of Labor. Consumer Price Index. Available at http://www.bls.gov/cpi/cpid1502.pdf Last accessed 14 January 2016.

ACKNOWLEDGMENTS

Editorial assistance was provided by Lisa Jolly and Katrina Rimmer of Caudex, UK, supported by Chimerix, Durham, NC.

DISCLOSURES

EM is an employee of Chimerix, Durham, NC. RC, LS, Y-CL, and VP are employees of LASER Analytica, New York, NY, which received research funds from Chimerix, Durham, NC, to conduct this database analysis, and for the development of this poster.

BMT Tandem Meetings; February 18–22, 2016; Honolulu, Hawaii