Infection Burden of Double Stranded DNA (dsDNA) Viruses after CD34+ Selected, T-cell Depleted (TCD) Hematopoietic Cell Transplantation (HCT) for Myeloid Malignancies at Memorial Sloan Kettering Cancer Center (MSK)

Y-T Huang[1], SJ Kim[1], YJ Lee[1,3], D Burack[1], P Nichols[1], M Maloy[2], R Tamari[2, 3], M-A Perales[2,3], S Giralt[2,3], H Castro-Malaspina[2,3], E Papadopoulos[2,3], A Jakubowski[2,3], G Papanicolaou[1,3]

[1]Infectious Disease Service, [2]Adult Bone Marrow Transplantation Service, Department of Medicine, Memorial Sloan Kettering Cancer Center, [3]Weill Medical College, Cornell University, New York, NY

Background

❖The ds DNA viruses Cytomegalovirus (CMV), Adenovirus (ADV), Human herpes virus 6 (HHV6) and Epstein-Barr virus (EBV) may cause serious infections in HCT recipients.

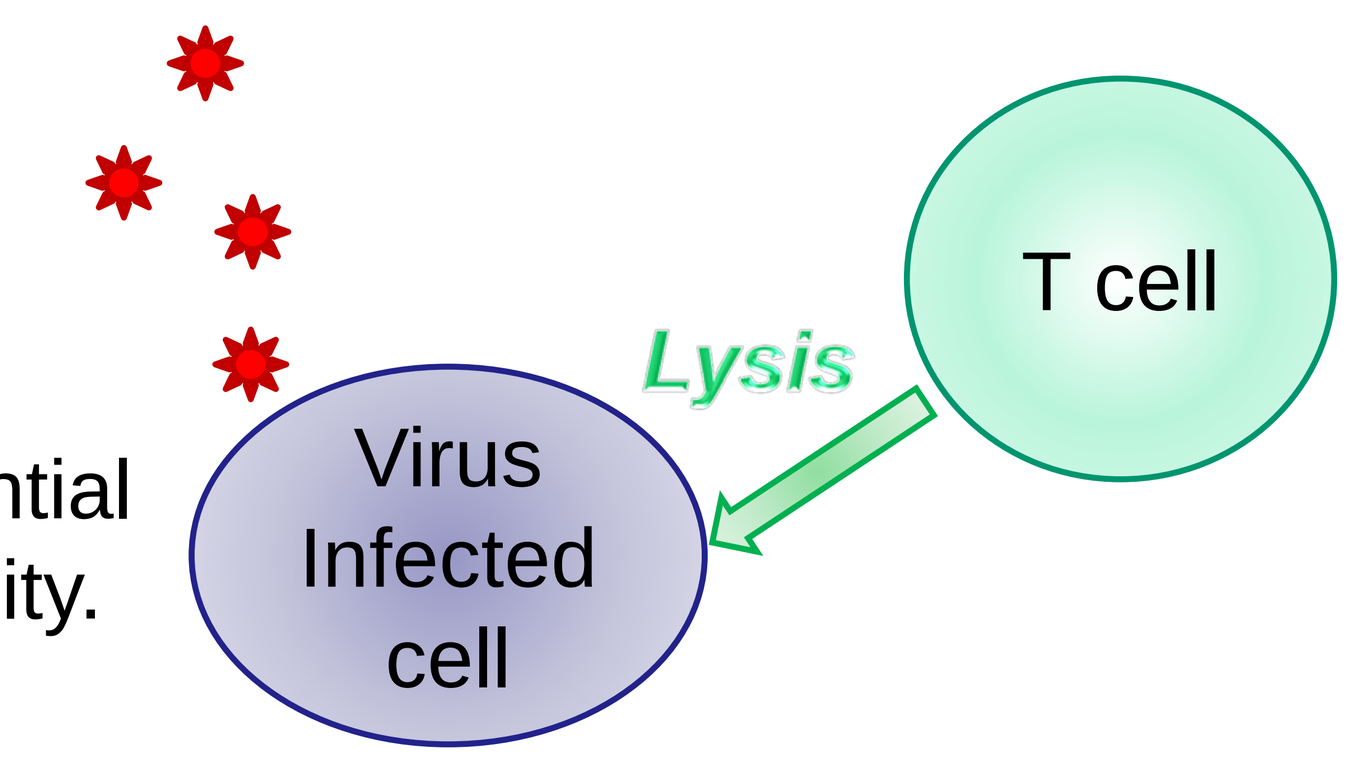
T cell
Lysis
Virus Infected cell

❖T-cells are essential for antiviral immunity.

❖A 5-log reduction of T-cells in the allograft is achieved by the Clinimacs® CD34+Selection System.

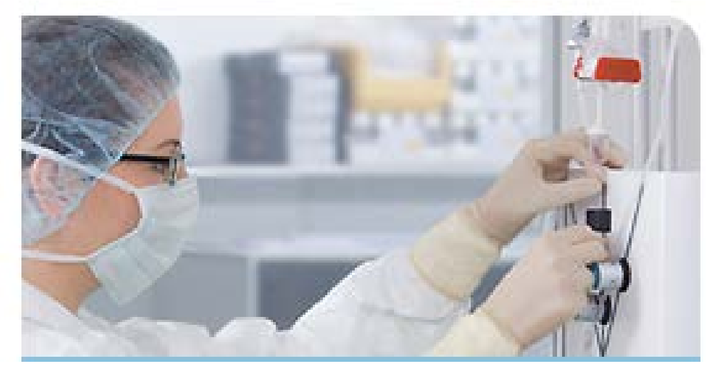

❖Recipients of *ex vivo* TCD HCT are susceptible to infections by DsDNA viruses.

Objectives

(1) Estimate the incidences of CMV, ADV, HHV6 and EBV viremia stratified by recipient CMV serostatus (R+ vs R-).

(2) Estimate the incidences of end organ disease (EOD) by CMV, ADV, HHV6 and EBV at 1 year post HCT.

(3) Estimate rates of co-infections by ds DNA viruses.

Methods

❖TCD HCT recipients of acute leukemia and myelodysplastic syndrome at MSK from June 2010 to December 2014 were routinely monitored by quantitative polymerase chain reaction (qPCR) assays for CMV, ADV, HHV6 and EBV.

❖End organ disease (EOD) was scored by standard criteria.

❖Cumulative incidence (CI) for viremia and EOD was estimated by the Kaplan-Meier method and compared by the log-rank test. *P*<0.05 was deemed statistically significant.

Rates of viremia and end organ disease (EOD)

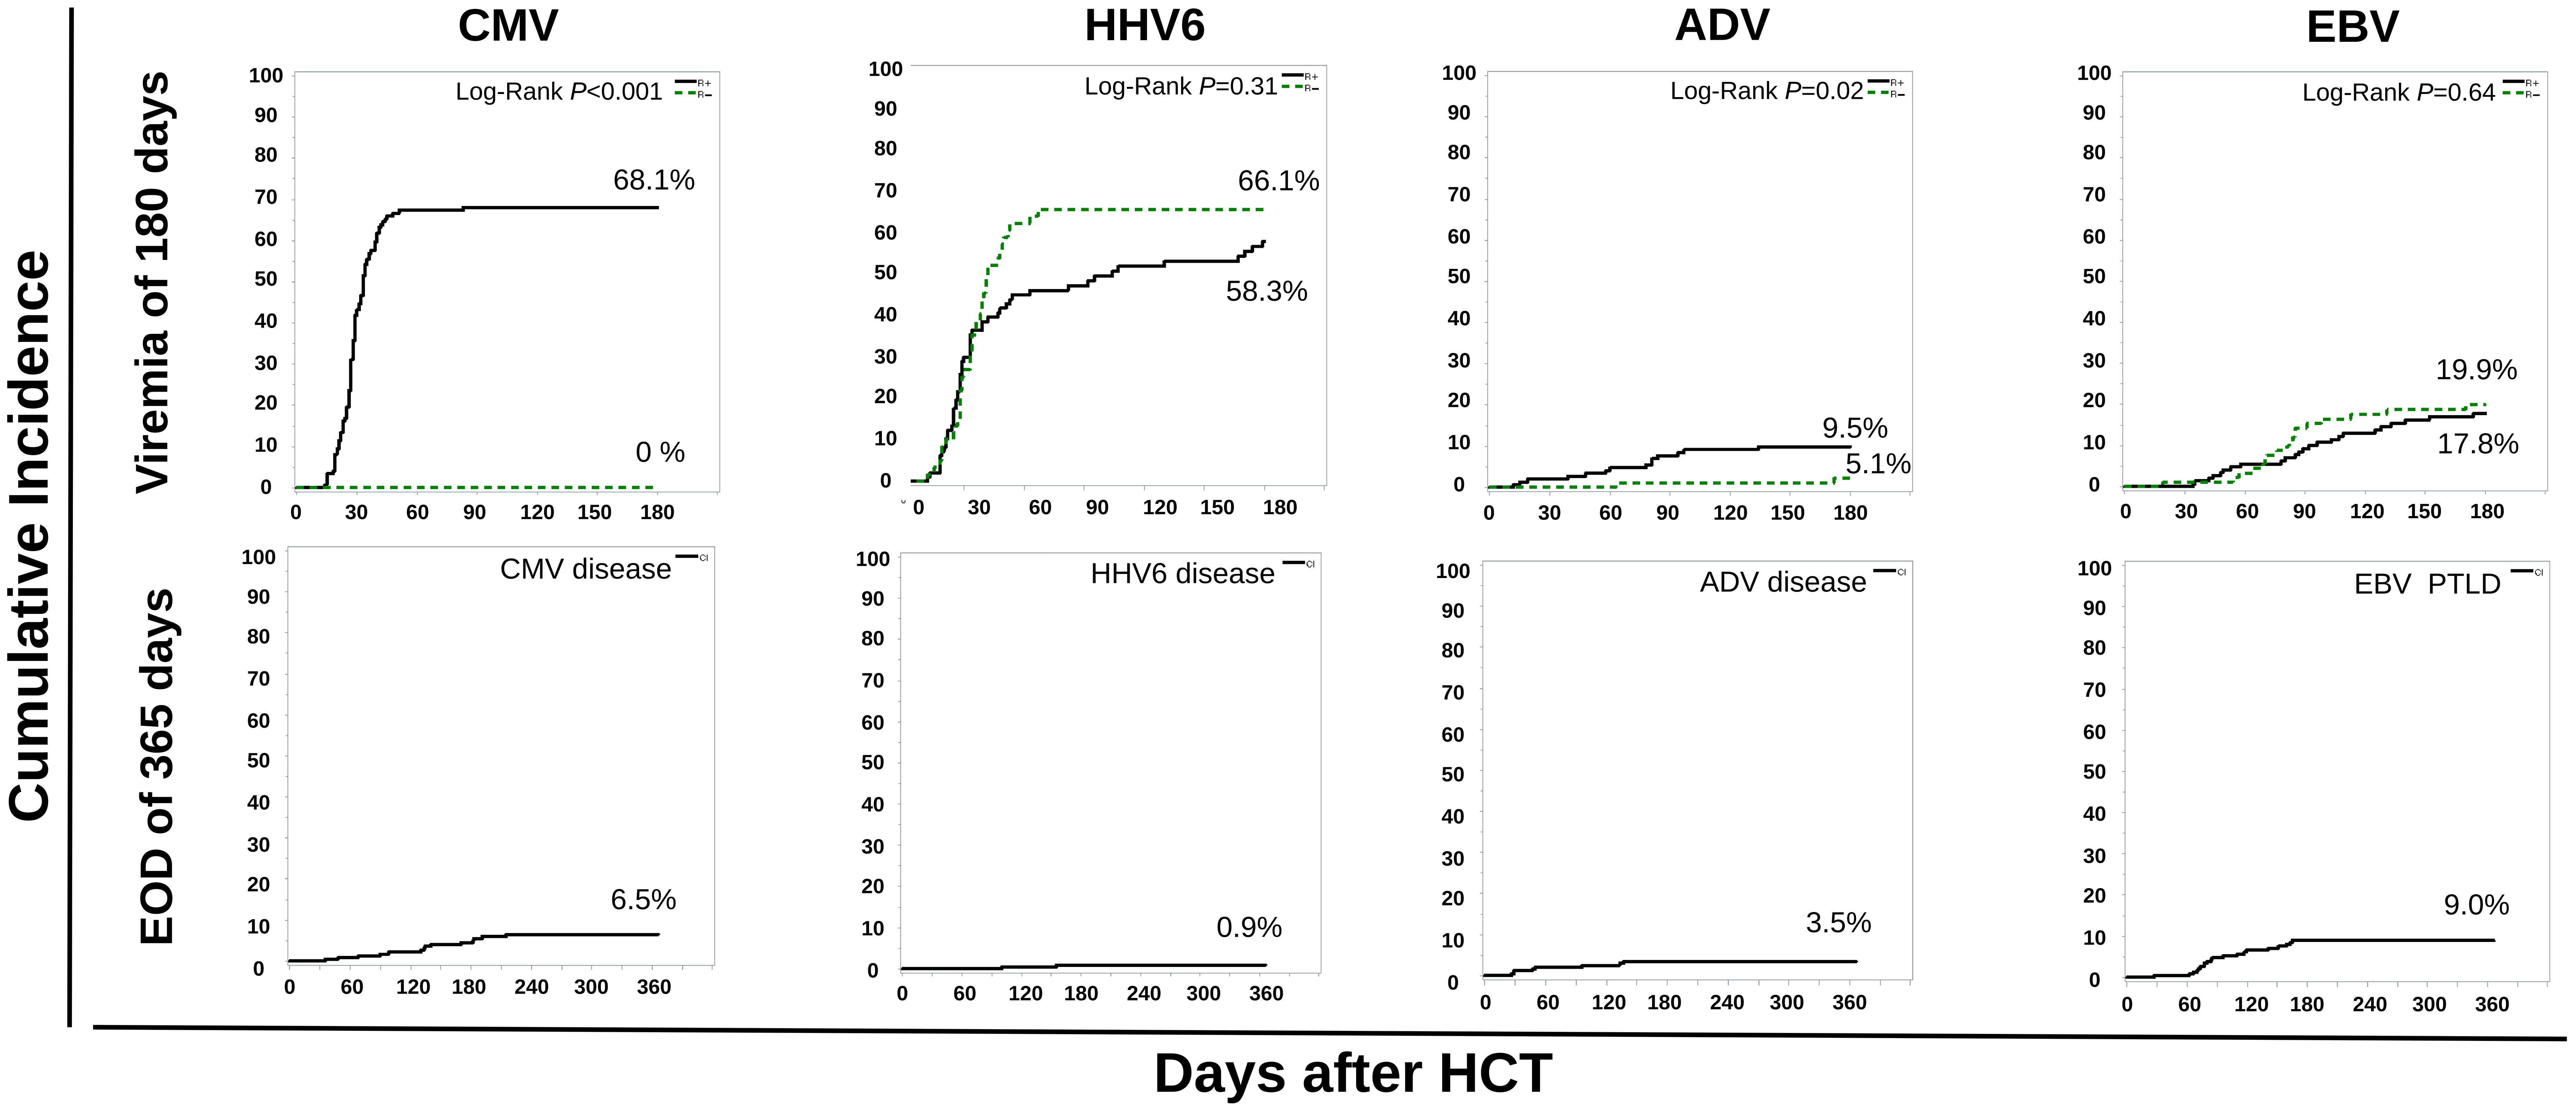
CMV
HHV6
ADV
EBV
Cumulative Incidence
Viremia of 180 days
EOD of 365 days
Log-Rank P<0.001
68.1%
0 %
Log-Rank P=0.31
66.1%
58.3%
Log-Rank P=0.02
9.5%
5.1%
Log-Rank P=0.64
19.9%
17.8%
CMV disease
6.5%
HHV6 disease
0.9%
ADV disease
3.5%
EBV PTLD
9.0%
0 30 60 90 120 150 180
0 60 120 180 240 300 360
Days after HCT

79% patients developed viremia and 77% had onset <100 days post HCT (N=239)

58% patients with CMV viremia had viral coinfections (N=100)

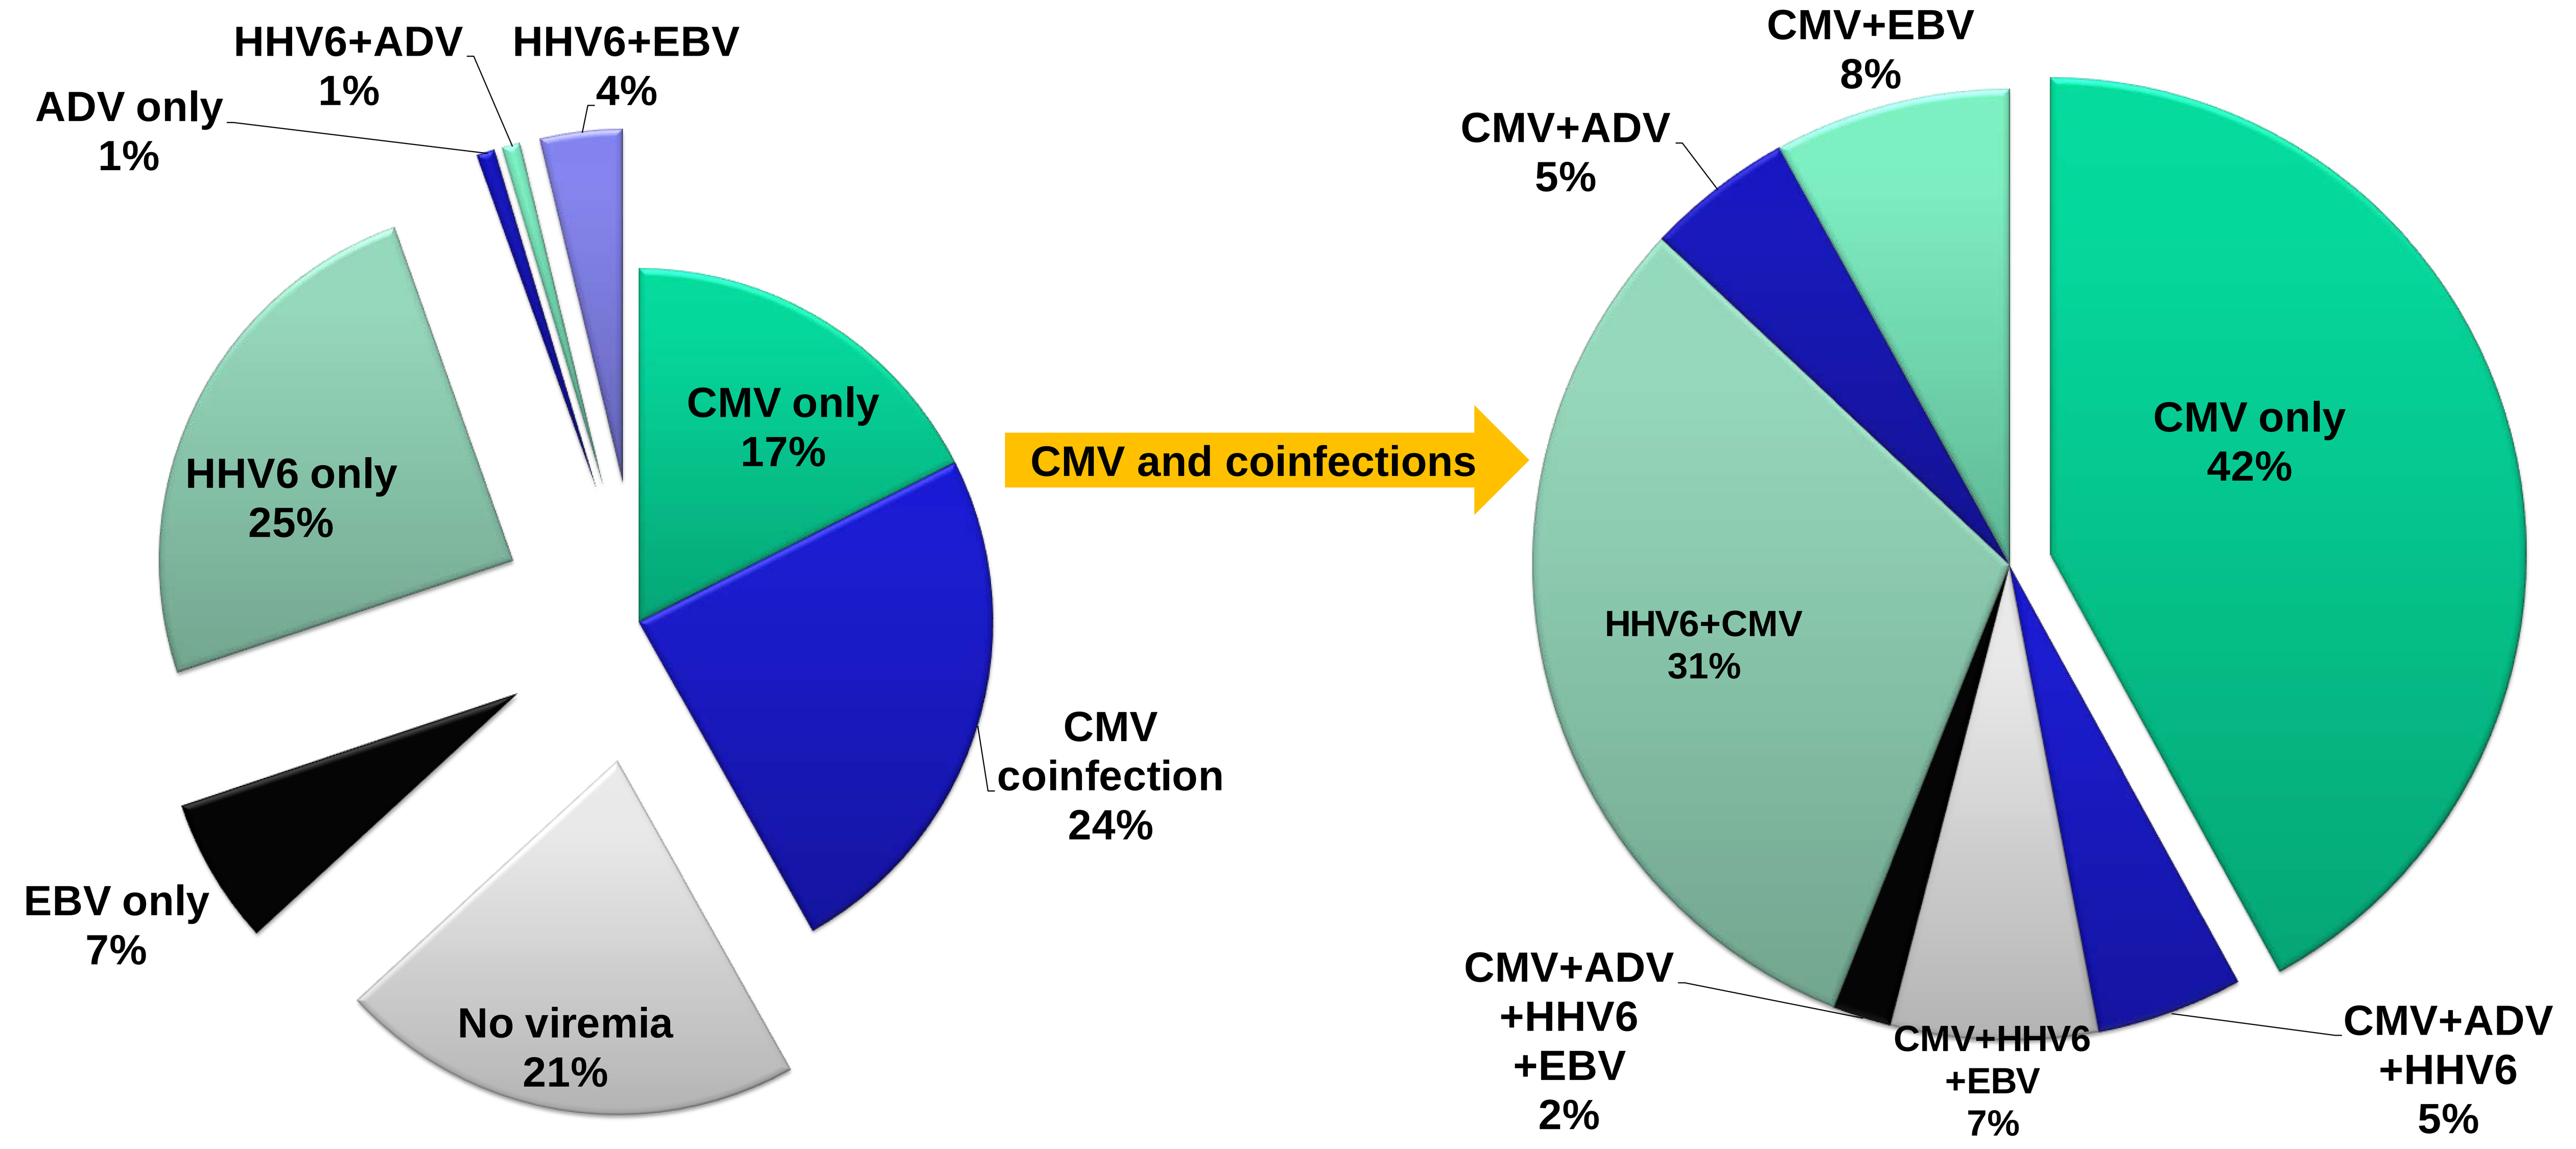
HHV6+ADV 1%
HHV6+EBV 4%
ADV only 1%
CMV only 17%
HHV6 only 25%
CMV coinfection 24%
EBV only 7%
No viremia 21%
CMV and coinfections
CMV+EBV 8%
CMV+ADV 5%
CMV only 42%
HHV6+CMV 31%
CMV+ADV +HHV6 +EBV 2%
CMV+HHV6 +EBV 7%
CMV+ADV +HHV6 5%

Conclusions

1) 77% of patients developed at least 1 viral infection, within the first 100 days post HCT.

2) Among patients with CMV viremia, 58% had coinfections with additional dsDNA virus(es).

3) Our data highlights the burden of viremias in recipients of TCD allografts.

4) Despite high rates of viremia, rates of end organ disease were low, supporting the effectiveness of active surveillance and aggressive preemptive therapy.

❖Acknowledgements: This research was funded in part through the NIH/NCI Cancer Center Support Grant P30 CA008748.

❖Contact information:
Genovefa A. Papanicolaou
papanicg@mskcc.org