Kidney Week
Philadelphia, PA
October, 2011

CMX001 was Well Tolerated and Without Nephrotoxicity in Renal Transplant and Hematopoietic Stem Cell Transplant Recipients with BK Virus Infection in a Prospective, Randomized, Double-Blind Study

V. Ram Peddi[1], Michael Boeckh[2], Steven A. Pergam[2], Eileen Scigliano[3], Deborah B. Adey[4], Daniel C. Brennan[5], Susan Godkin[6], Alice T. Robertson[6] and Wendy Painter[6]

[1]California Pacific Medical Center, San Francisco, CA, [2]Fred Hutchinson Cancer Research Center, Seattle, WA, [3]Mount Sinai Medical Center, New York, New York, [4]University of Vermont, Burlington, VT, [5]Washington University in Saint Louis, St. Louis, MO and [6]Chimerix Inc., Durham, NC, USA

BACKGROUND

CMX001 is an oral lipid-antiviral-conjugate that produces high intracellular levels of the active antiviral agent cidofovir (CDV)-diphosphate and has potent in vitro activity against all five families of dsDNA viruses that affect humans including polyomaviruses such as BK virus (BKV), herpesviruses such as cytomegalovirus (CMV), papillomaviruses, adenoviruses and orthopoxviruses, . Non-clinical studies with CMX001 have shown no evidence of CDV-like, dose-limiting nephrotoxicity. Unlike CDV, CMX001 is not a substrate of human Organic Anion Transporter 1 (hOAT1) (1), thereby providing a mechanistic explanation for the lack of CDV-like nephrotoxicity following administration of CMX001.

BKV can cause significant disease in immunosuppressed patients such as BKV associated nephropathy (BKVAN) in renal transplant (RT) recipients and hemorrhagic cystitis (HC) in hematopoietic stem cell transplant (HSCT) recipients. BKVAN can result in allograft dysfunction or graft loss in RT recipients and is often managed by a reduction in immunosuppression which can increase the risk of graft rejection. HC can be an extremely painful and debilitating condition which may prolong hospitalization, require supportive care including transfusions and result in impairment of kidney function in HSCT recipients. There are currently no approved therapies for BKV infection and the side effects associated with some of the most commonly used unproven agents are problematic and can be dose-limiting. CMX001 has the potential to offer a treatment option for this unmet medical need.

The safety and tolerability of CMX001, including its effect on renal function, was evaluated in CMX001-104, a safety and tolerability study in RT and HSCT recipients with BKV infection.

METHODS

Adult RT and HSCT recipients with BKV infection were randomized to CMX001 40mg or placebo (2:1) once weekly for a total of 5 doses.

Key qualifying criteria for RT recipients were:
1. **At least 28 days post-transplant**
2. **Hemoglobin > 10g/100mL**
3. **Stable immunosuppression for 14 days prior to dosing**
4. **Either**
 a) **BKV viruria ≥ 10^4 copies/mL without viremia or**
 b) **BKV viremia <10^4 copies/mL with or without viruria**

Key qualifying criteria for HSCT recipients were:
1. **A minimum of 3 days post successful engraftment with an ANC>500 cells/mm^3**
2. **BKV viruria ≥ 10^4 copies/mL**

•All subjects must have had a GFR > 30mL/min

Subjects were not eligible if they met any of the following criteria:
•Had received any medication for treatment of BKV infection or disease within 14 days prior to enrollment
•Had a bilirubin level > 2.5 X ULN
•Had concurrent or ongoing ≥ Grade 2 GI symptoms

The study included a 35 day Treatment Period and a 28 day Follow-up period. Clinical safety labs, viral load and adverse event assessments were performed and blood samples were collected for PK analysis.

Subjects were to have been discontinued from the Treatment Phase if they experienced a Grade 4 drug-related AE or laboratory abnormality, their GFR decreased by ≥ 50% or to <20mL/min, they developed CMV infection requiring treatment with an excluded medication, or they became pregnant. Additionally, RT recipients were excluded from the Treatment Phase if they had a significant change in their immunosuppressant therapy.

The following medications were prohibited throughout the study:
•Cidofovir
•Leflunomide
•Any investigational medication

During the Treatment Phase, IV aminoglycosides and NSAIDs (other than cardioprotective) were prohibited and fluoroquinolones were discouraged; valganciclovir and ganciclovir were prohibited for HSCT recipients, due to their myelosuppressive properties.

RESULTS

12 RT recipients and 11 HSCT recipients were enrolled and all completed the Treatment Phase of the study. The demographics and baseline BKV viral loads of these subjects are displayed in Table 1 below.

Table 1: Demographics and baseline BKV viral loads

	RT CMX001 (n=8)	RT Placebo (n=4)	HSCT CMX001 (n=7)	HSCT Placebo (n=4)
Age (years) Median (range)	58 (28-76)	50 (45-72)	49 (36-68)	33.5 (18-59)
Gender	3 Female; 5 Male	1 Female; 3 Male	7 Male	3 Female; 1 Male
Race	7 White; 1 Black	4 White	4 White;2 Black; 1 Hispanic	4 White
BMI (kg/m2) Median (range)	26.9 (22.8-31.8)	23.5 (20.3-41.1)	25.2 (19.3-31.0)	23.8 (22.2-27.2)
Baseline BKV viruria Mean (SD) log copies/mL	6.51 (1.40)	7.39 (1.74)	6.56 (1.85)	9.00 (1.54)
Subjects with BKV viremia >500 log copies/mL at baseline, N (%)	3 (37.5)	0	0	2 (50.0)

The transplant histories of the RT and HSCT subjects are shown in Tables 2 and 3, respectively.

Table 2: RT subjects transplant histories

	RT CMX001 (n=8)	RT Placebo (n=4)
Days from transplant to first dose Median (range)	614.5 (125-2490)	746.5 (163-1044)
Kidney source	6 cadaver 2 living donor	3 cadaver 1 living donor

Table 3: HSCT subjects transplant histories

	HSCT CMX001 (n=7)	HSCT Placebo (n=4)
Days from transplant to first dose Median (range)	121 (48-566)	74 (52-112)
Stem cell source (all were allogeneic)	1 cord blood 6 peripheral blood	1 bone marrow 1 cord blood 2 peripheral blood

Safety and Adverse Events

There were no drug-related SAEs or remarkable AEs and none of the subjects discontinued from the study due to an adverse event.

Renal function was monitored via clinical lab results, including GFR which was calculated using the MDRD formula. Mean GFR, serum creatinine and BUN at baseline and Day 35 (7 days after the last dose of study drug) are shown in Table 4.

Table 4: Mean (SD) GFR, Serum Creatinine and BUN

	RT CMX001 (n=8)	RT Placebo (n=4)	HSCT CMX001 (n=7)	HSCT Placebo (n=4)
GFR (mL/min/1.73m^2)				
Day 0/Baseline	55.5 (11.1)	49.8 (8.3)	83.0 (31.2)	66.5 (20.2)
Day 35	54.4 (10.5)	44.3 (4.7)	79.9 (32.3)	69.8 (40.5)
Change from Baseline	-1.1 (2.8)	-5.5 (4.8)	-3.1 (29.2)	3.3 (32.1)
Serum Creatinine (mg/dL)				
Day 0/Baseline	1.29 (0.27)	1.43 (0.28)	1.11 (0.38)	1.13 (0.40)
Day 35	1.31 (0.27)	1.55 (0.19)	1.13 (0.28)	1.18 (0.51)
Change from Baseline	0.03 (0.07)	0.13 (0.10)	0.01 (0.31)	0.05 (0.26)
BUN (mg/dL)				
Day 0/Baseline	20.9 (5.44)	19.0 (6.98)	21.9 (8.88)	25.0 (4.69)
Day 35	22.4 (3.89)	20.8 (8.96)	18.0 (11.11)	21.5 (11.47)
Change from Baseline	1.5 (7.01)	1.8 (3.30)	-3.9 (5.61)	-3.5 (10.66)

Additionally, there were no significant changes in proteinuria. Changes in ANC and LFTs were evaluated and the results are presented in Tables 5 and 6, respectively.

Table 5: Mean (SD) Absolute Neutrophil Count (K/mm^3)

	RT CMX001 (n=8)	RT Placebo (n=4)	HSCT CMX001 (n=7)	HSCT Placebo (n=4)
Day 0/Baseline	3.56 (1.00)	3.65 (1.86)	5.10 (2.42)	7.40 (5.90)
Day 35	3.93 (1.44)	3.70 (1.65)	3.79 (2.48)	1.90 (1.27)
Change from Baseline	0.08 (1.13)	0.05 (0.42)	-1.19 (0.99)	-5.3 (4.30)

Table 6: Mean (SD) ALT, AST, Alkaline Phosphatase and Total Bilirubin

	RT CMX001 (n=8)	RT Placebo (n=4)	HSCT CMX001 (n=7)	HSCT Placebo (n=4)
ALT (u/L)				
Day 0/Baseline	19.5 (7.6)	18.8 (11.2)	36.4 (26.9)	37.5 (12.6)
Day 35	22.9 (12.4)	18.5 (9.3)	29.3 (18.5)	24.0 (12.4)
Change from Baseline	3.4 (5.2)	-0.3 (2.5)	-7.1 (14.9)	-13.5 (11.0)
AST (u/L)				
Day 0/Baseline	20.0 (4.7)	21.8 (3.1)	24.7 (8.4)	27.5 (8.7)
Day 35	22.1 (7.0)	22.3 (6.2)	23.0 (7.2)	21.5 (9.0)
Change from Baseline	2.1 (4.9)	0.5 (3.7)	-1.7 (9.2)	-6.0 (5.0)
Alk Phos (u/L)				
Day 0/Baseline	86.8 (17.6)	102.8 (54.0)	70.4 (38.3)	82.3 (12.9)
Day 35	92.3 (15.6)	97.8 (40.0)	66.7 (23.6)	84.3 (25.6)
Change from Baseline	5.5 (11.8)	-5.0 (14.2)	-3.7 (18.8)	2.0 (21.6)
Total Bilirubin (mg/dL)				
Day 0/Baseline	0.34 (0.13)	0.48 (0.36)	0.74 (0.39)	0.75 (0.33)
Day 35	0.46 (0.26)	0.46 (0.26)	0.90 (0.97)	0.44 (0.10)
Change from Baseline	0.12 (0.16)	-0.02 (0.11)	0.16 (0.71)	-0.30 (0.31)

CMX001 Concentration-time Profile

CMX001 mean concentrations in RT and HSCT recipients following a 40mg oral dose are shown below.

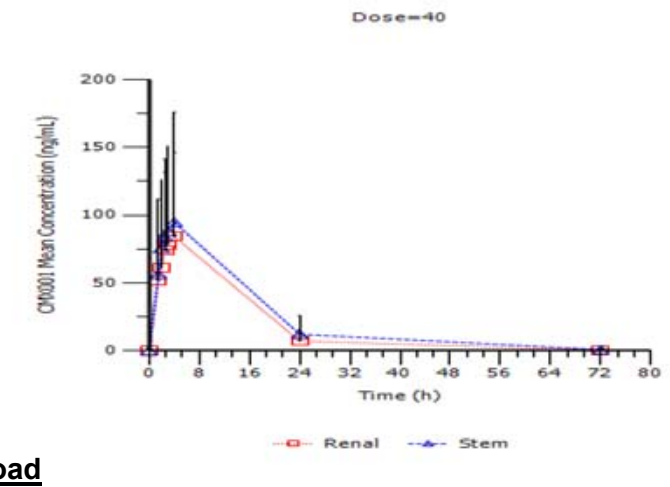
Dose=40
CMX001 Mean Concentration (ng/mL)
200
150
100
50
0
0
8
16
24
32
40
48
56
64
72
80
Time (h)
Renal
Stem

Viral Load

While the sample size in this study was not sufficient to assess the effectiveness of CMX001 in treating BKV infection, the results, as shown in Table 7 below, support further evaluation.

Table 7: Number of subjects (%) experiencing changes in viral load

	RT CMX001 (n=8)	RT Placebo (n=4)	HSCT CMX001 (n=7)	HSCT Placebo (n=4)
2-log drop in BKV viruria (%)	2 (25)	0	3 (42.9)	1 (25)
Cleared BKV viruria (%)	1 (12.5)	0	1 (14.3)	0
Cleared baseline BKV viremia	2 of 3 (67%)	N/A	N/A	1 of 2 (50%)
Emergence of or a 2-log increase in BKV viremia	0	0	0	1 (25%)

Implications for Future Studies

No dose-limiting toxicities were identified during this study; thus, the data support investigation of higher doses of CMX001 in these patient populations. Ongoing studies with CMX001 in RT and HSCT subjects are evaluating total weekly doses of up to 200mg in adults.

CONCLUSIONS

1. **Five weekly oral doses of CMX001 40mg in RT and HSCT recipients with BKV infection were well tolerated without adverse renal effects. This is consistent with results from other CMX001 clinical trials and experience with subjects treated under EIND (2).**
2. **No clinically significant changes in GFR, proteinuria, ANC, ALT, AST, Alk Phos or bilirubin were observed during this study.**
3. **The concentration-time profiles of CMX001 observed in this study, following a single 40mg oral dose, do not appear different between RT and HSCT recipients.**
4. **While this study is small, the viral load results support further evaluation of CMX001 for prevention and/or treatment of BKV infection. Further evaluation of CMX001 as an anti-BKV therapy is planned.**

REFERENCES

1. Tippin TK et al., (2010) American Association of Pharmaceutical Scientists
2. Papanicolaou G et al. (2011) 51St Interscience Conference on Antimicrobial Agents and Chemotherapy

CHIMERIX, INC.