Poster #502

BMT Tandem Meetings February 2011

Experience with CMX001, a Novel Antiviral Drug for Cytomegalovirus Infections in Stem Cell Transplant Patients

Genovefa Papanicolaou, MD[1], Joanne Kurtzberg, MD[2], Peter Westervelt, MD[3], Juan Gea-Banacloche, MD[4], Erica Warlick, MD[5], Randall Lanier, PhD[6], Maggie Anderson[6], and Wendy Painter, MD, MPH[6]

[1]Memorial Sloan Kettering Cancer Center, [2]Duke University Medical Center, [3]Washington University School of Medicine, [4]National Cancer Institute, [5]University of Minnesota, [6]Chimerix, Inc.

Abstract

Title: Experience with CMX001, a Novel Antiviral Drug, for Cytomegalovirus Infections in Stem Cell Transplant Patients

Genovefa Papanicolau, MD[1], Joanne Kurtzberg, MD[2], Peter Westervelt, MD[3], Juan Gea_Banacloche, MD[4], Erica Warlick, MD[5], Randall Lanier, PhD[6], Maggie Anderson[6] and Wendy Painter, MD, MPH[6]. [1]Memorial Sloan Kettering Cancer Center, New York, NY, United States; [2]Pediatric Blood and Marrow Transplantation, Duke University Medical Center, Durham, NC, United States; [3]Department of Medicine Oncology, Washington University School of Medicine, St. Louis, MO, United States; [4]Experimental Transplantation and Immunology Branch, National Cancer Institute, Bethesda, MD, United States; [5]Division of Hematology, Oncology, and Transplantation, University of Minnesota, Minneapolis, MN, United States and [6]Chimerix, Inc., Durham, NC, United States.

Body: Cytomegalovirus (CMV) infections are associated with significant morbidity and mortality in the stem cell transplant setting. CMX001, a lipid conjugate of cidofovir is administered orally and circulates as the lipid conjugate in plasma; it is efficiently taken up by target cells and high concentrations of the active antiviral are achieved intracellularly. We describe the first clinical experiences in stem cell transplant patients infected with CMV who received CMX001. The patients had history of AML (3 patients), refractory lymphoma, multiple myeloma, and severe aplastic anemia, and sickle cell anemia. Six of the seven patients had received SCT and the seventh awaited SCT. Treatment with CMX001 was initiated pre-transplant in one patient, and 21 days to greater than 2 years in six patients (median of 61days).

We analyzed 7 patients with CMV viremia who were treated under emergency IND because of lack of other reasonable therapeutic options. All 7 patients had failed conventional antiviral therapy: all patients had received ganciclovir and/or valganciclovir, 6 had received foscarnet, and 4 had received CMV IG. The doses of CMX001 in these patients ranged from 80 mg to 300 mg (approximately 2 to 4 mg/kg); follow-up data was available for at least 4 weeks in all patients. Virologic response was defined as more than a 90% reduction (1 log 10) in viral load (VL) and complete response was defined as an undetectable viral load.

The 4 males and 3 females treated had a median age of 55 years (range 11 to 69 years); they were treated with CMX001 for a median of 88 days (range 29-131 days). The median reduction in VL was greater than 1.2 log 10 at 4 weeks. A complete response was observed in 3/5 (60%) patients who did not have mutations in the CMV polymerase UL54 gene; 2/5 had an average reduction in CMV by PCR of 1.2 log 10. Neither of two patients with a relevant mutation in UL54 (L501F and A987G) had a 1 log reduction in viremia at the last time point.

Two patients had pre-existing renal insufficiency; no dose adjustment was needed based on kidney function. During treatment with CMX001, one patient experienced C. difficile colitis, one experienced pancytopenia along with GVHD and sepsis, one with a seizure disorder experienced seizure, and one experienced severe GVHD. No trends were observed in SAEs.

This case series demonstrates that CMX001 is a promising therapeutic option for the treatment of CMV disease in stem cell transplant patients.

Background

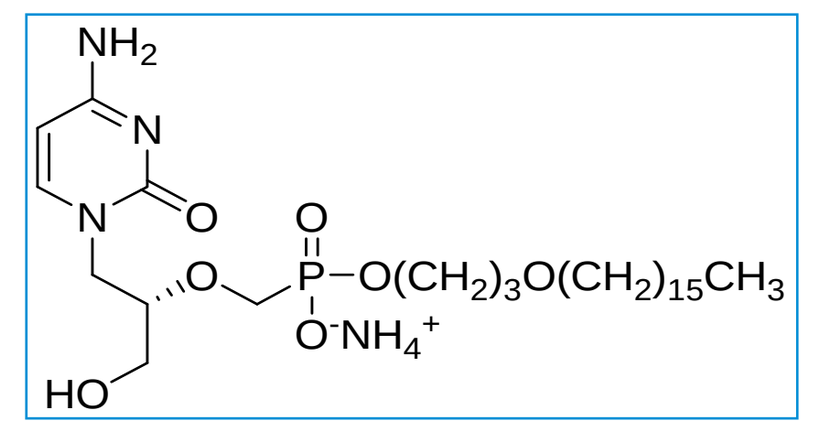

CMX001 is an orally administered antiviral agent which is absorbed in the small intestine, delivered to target organs, cleaved by intracellular phospholipases, and then converted to the active antiviral agent, CDV-diphosphate. Compared to CDV, CMX001 has significantly higher *in vitro* antiviral activity against double-stranded DNA (dsDNA) viruses of clinical importance to the transplant community including herpesviruses, adenoviruses, and polyomaviruses.

Early in the development program for CMX001, an unmet medical need was identified in immunocompromised patients with dsDNA virus infections who had no other options for antiviral therapy. Physician-held Emergency INDs (EINDs) were granted by the FDA and comparable ex-US regulatory authorities allowing for treatment of individual patients with CMX001. To qualify for treatment, patients must have had an immediately life-threatening condition for which there were no other therapeutic options. Since March 2009, over 150 EINDs have been granted for treatment of pediatric or adult patients; of these, 33 patients (12 pediatric and 21 adult) were being treated as of January 2011.

In this poster, we present case studies of 7 stem cell transplant recipients treated under EINDs in the US. All patients had refractory CMV, multiple comorbidities, and limited life-expectancy. All EIND patients who met the following criteria as of 31 August 2010 were selected for presentation: 1) received or awaiting stem cell transplant, 2) CMV was the primary viral infection, 3) patient had failed all other anti-CMV therapies, 4) patient had been treated with CMX001 for a minimum of 4 weeks and virology data was available, 5) CMV viremia was greater than 1000 copies/mL within 7 days prior to initiation of CMX001 therapy. None of these cases are ongoing (under an EIND) at this time.

Case Study #1

Medical History: The patient was a 28 year old female with refractory angioimmunoblastic T-cell lymphoma who was receiving salvage chemotherapy with mini-Beam at time of request for CMX001. The patient had evidence of disease progression noted in the bone marrow, new bilateral, likely-malignant lung nodules as well as liver and lymph node progression detected on CT scan.

Reason for EIND/request for CMX001: The patient had ongoing CMV viremia that was resistant to GCV (UL97 mutation detected) and was persistent despite foscarnet therapy. The patient needed to clear CMV to be eligible to receive a cord blood transplant which was planned for 5 to 6 weeks later.

Treatment Emergent Comorbidities: Soon after initiation of CMX001 therapy, blood culture was positive for *P. aeruginosa*, and urine culture was positive for *K. pneumoniae* and *C. freundie*. The patient also developed BK virus associated hemorrhagic cystitis which resolved during treatment.

CMV Infection and Treatment: Due to persistent CMV viremia, CMX001 was initiated at 2 mg/kg twice weekly. While receiving CMX001, the patient developed CMV retinitis which necessitated re-initiation of foscarnet therapy. On combination therapy, CMV retinitis was controlled and CMV viremia decreased.

CMX001 Therapy			CMV Viremia			
Dose (mg) Initial / Final	Timing of First Dose (Months post-transplant)	Duration of Treatment	Baseline (copies/mL)	End of therapy (copies/mL)	Reduction (log_{10})	Comments
120 / 120	pre-transplant	127 days	5000	1500 450	-0.52 -1.05	3 wks monotherapy; dev. CMV retinitis added foscarnet; controlled retinitis

Case Study #2

Medical History: The patient was a 62 year old female with a history of MDS with progression to AML who underwent a matched unrelated donor stem cell transplant in 2009. Patient had gut GVHD and eventually withdrew treatment due to acute renal failure secondary to severe gut GVHD.

Reason for EIND/request for CMX001: The patient had ongoing CMV viremia that was resistant to GCV and increasing viral loads while on foscarnet/cytogam therapy.

CMV Infection and Treatment: Following transplant, the patient had multiple recurrences of CMV viremia. She was treated with GCV, switched to foscarnet, and then cytogam was added to the foscarnet regimen but her CMV continued to rise. Genetic testing confirmed the presence of a UL97 mutation. Treatment with CMX001 (3 mg/kg twice weekly) resulted in reduction in plasma CMV viral load. CMV enteritis was present at the end of the treatment period

CMX001 Therapy			CMV Viremia		
Dose (mg) Initial / Final	Timing of First Dose (Months post-transplant)	Duration of Treatment	Baseline (copies/mL)	End of therapy (copies/mL)	Reduction (log_{10})
180 / 180	5 months	38 days	255,485	4,339	-1.77

Case Study #3

Medical History: The patient was a 55 year old male who underwent a haploidentical allogeneic stem cell transplant in April 2009 for AML. He had poor immunoreconstitution with lack of T cell recovery and persistent CMV viremia one year following transplant. He was given haploidentical donor lymphocytes to provide some immune reconstitution, but developed severe GI GVHD that was not controlled by standard treatments. Comorbidities included encephalitis, meningoencephalitis (appeared to be CMV), and numerous complex cysts within both kidneys. The patient also had many other infectious complications including rhodococcal bacteremia, rhizopus lung infection, and persistent RSV pneumonia.

Reason for EIND/request for CMX001: The patient had CMV with mutations conferring resistance to GCV and CDV, and was intolerant to foscarnet.

Treatment Emergent Comorbidities: Emergent events included pancytopenia, renal failure, hypernatremia, dehydration, abdominal pain, and diarrhea.

CMV Infection and Treatment: The patient had persistent viremia, and biopsy confirmed CMV involvement in the GI track. Prior to initiating therapy, CMV with a UL54 mutation conferring resistance to CDV had been detected. Treatment with CMX001 (4 mg/kg twice weekly) did not result in a significant drop in CMV viremia.

CMX001 Therapy			CMV Viremia			
Dose (mg) Initial / Final	Timing of First Dose (Months post-transplant)	Duration of Treatment	Baseline (copies/mL)	End of therapy (copies/mL)	Reduction (log_{10})	Comments
260 / 260	12 months	36 days	9200	3500	-0.42	UL54 mutation

Case Study #4

Medical History: The patient was an 11 year old female with multiple relapsed AML and refractory CMV who had received a T-cell depleted HLA matched related allogeneic stem cell transplant. She had a refractory bacteremia and had an ileostomy as a result of a previous fungal infection. The patient had a positive blood culture for gram positive cocci, asymptomatic BKV viruria, and had tested positive for AdV in stool samples. The subject did not have evidence of GVHD.

Reason for EIND/request for CMX001: The patient had been treated with foscarnet but experienced continually rising antigenemia, so GCV was added to her regimen. Following addition of GCV, the patient experienced a drop in WBC/ANC so GCV treatment was halted. Concern that foscarnet was not effective as monotherapy and may be overly toxic to the kidneys resulted in the request for an EIND.

Treatment Emergent Comorbidities: The patient developed pneumonia which resolved while on treatment.

CMV Infection and Treatment: The patient was CMV positive based on antigenemia and plasma PCR results. There was no evidence of CMV disease. The subject responded to CMX001 treatment (3 mg/kg twice weekly) as CMV viremia became and remained negative throughout treatment.

CMX001 Therapy			CMV Viremia	
Dose (mg) Initial / Final	Timing of First Dose (Months post-transplant)	Duration of Treatment	Baseline (copies/mL)	End of therapy (copies/mL)
80 / 80	1.5 months	181 days	1000	undetectable

Case Study #5

Medical History: The patient was a 25 year old male with refractory CMV. He had sickle cell anemia with multiple complications including strokes, cholelithiasis s/p cholecystectomy, multiple pain crises, renal insufficiency and chest syndromes as well as a history of seizure disorders. He had received a cord blood transplant in 2001 and haploidentical transplant in Jan. 2010.

Reason for EIND/request for CMX001: Patient had failed to engraft following 2 previous transplants and was scheduled for a third transplant in a few months; however, the patient had reactivated CMV and had rising viremia while on GCV which put scheduling of the next transplant in jeopardy.

Treatment Emergent Comorbidities: The physician reported a serious event of persistent diarrhea and readmission to the hospital due to seizure (both considered not related to CMX001). In addition, CMV colitis and transient transaminitis were reported.

CMV Infection and Treatment: The patient had CMV viremia that had responded initially to iv GCV. He had been switched back and forth from iv GCV to vGCV and cytogam had been added at various times in an attempt to control his CMV viremia. The patient had a UL54 mutation, but had initially responded to combination therapy with CMX001 and iv GCV.

CMX001 Therapy			CMV Viremia			
Dose (mg) Initial / Final	Timing of First Dose (Months post-transplant)	Duration of Treatment	Baseline (copies/mL)	End of therapy (copies/mL)	Reduction (log_{10})	Comments
300 / 150	5 months	88 days	18,430	13,460 4226	-0.14 -0.64	2 wks monotherapy, UL54 mt. +GCV combination

Case Study #6

Medical History: The patient was a 69 year old male with refractory multiple myeloma who had previously received an autologous SCT and allogenic BMT. Most recently, he had received a T-cell depleted PBSCT from a matched related donor. The patient's course had been complicated by recurrent CMV, BK virus hemorrhagic cystitis, and disseminated zoster.

Reason for EIND Request for CMX001: CMX001 was requested due to the patient's persistent CMV reactivation, disseminated VZV which developed while the patient was on foscarnet maintenance for CMV and acyclovir for zoster, and a history of Valcyte-related neutropenia.

Treatment Emergent Comorbidities: During treatment with CMX001, the patient developed *C. difficile* colitis which later resolved.

CMV Infection and Treatment: The patient had 2 courses of therapy with CMX001 (both were 200 mg twice weekly). After 36 days of initial therapy with CMX001 followed by 9 weeks off treatment, CMX001 was restarted due to reactivation of CMV viremia. CMV viremia cleared after 3 weeks of initial therapy. VZV lesions cleared during treatment. During the 2nd treatment course, CMV viremia cleared after 2 weeks of treatment.

CMX001 Therapy			CMV Viremia	
Dose (mg) Initial / Final	Timing of First Dose (Months post-transplant)	Duration of Treatment	Baseline (copies/mL)	End of therapy (copies/mL)
200 / 200	6 months	36 days	500	undetectable
9 weeks without CMX001 treatment				
200 / 200	9 months	22 days	12,150	undetectable

Case Study #7

Medical History: The patient was a 58 year old male with refractory relapsed severe aplastic anemia who received a bone marrow transplant from a matched related donor. His course was complicated by GVHD of the gut.

Reason for EIND/request for CMX001: The patient has late reactivation CMV refractory to both foscarnet and CMV IgG, and resistant to GCV. His treatment was complicated by worsening renal failure due to foscarnet.

Comorbidities: The investigator reported a case of severe pancytopenia which was attributed to GVHD, sepsis and medication including possibly CMX001. The patient also had BKV viruria.

CMV infection and treatment: The patient had viremia caused by CMV resistant to GCV and foscarnet. Following treatment with CMX001, CMV viral loads became undetectable.

CMX001 Therapy			CMV Viremia		
Dose (mg) Initial / Final	Timing of First Dose (Months post-transplant)	Duration of Treatment	Baseline (copies/mL)	End of therapy (copies/mL)	Comments
200 / 200	5 months	32 days	1604	undetectable	UL54 mt conferring resistance to GCV and foscarnet

Results:

Overview and Safety: We have presented case summaries from seven patients with life-threatening conditions who were treated with CMX001 for CMV under emergency use. The seven patients included 4 male and 2 female adult patients (median age 56.5) and one female pediatric patient (11 years of age). Cases 1 – 5 initiated dosing prior to June 2010 when the CMX001 dose was adjusted based on weight; subsequently, pharmacokinetic data has indicated that CMX001 exposure is not correlated to body weight in adults so the last 2 cases were treated using a regimen of 200 mg twice weekly. Three patients were treated for approximately 1 month (32, 36, and 38 days) and one patient was treated with 2 courses approximately 1 month in length (36 and 22 days). The duration of treatment for the 3 other patients ranged from 3 to 6 months (88, 127, and 181 days). Two patients had pre-existing renal insufficiency prior to dosing, neither of which required dose adjustment of CMX001. There were no trends in the type or intensity of serious adverse events reported using the MedWatch system.

Virologic Response: CMV response to CMX001 therapy appeared to correlate with the presence or absence of mutations in the viral polymerase (UL54) gene, as expected. Three patients had a complete response (i.e., CMV viremia was not detectable by PCR) to CMX001 therapy (Cases 4, 6, and 7). One case (#2) had a reduction in CMV viremia of 1.77 log_{10} but plasma CMV DNA did not become undetectable by PCR before CMX001 was withdrawn. In the 2 patients with mutations in the CMV DNA polymerase gene which are associated with CDV resistance, response to CMX001 was attenuated. Combination antiviral therapy was administered in two cases.

Pt.	Overall Response	Clinical Resistance / Mutation	Comment
Significant Virologic Response			
2	-1.77 log_{10}	Resistant to GCV / UL97	
4	to undetectable	None reported	
6		None reported	
7		Resistant to FOS, GCV / mutations not reported	
Combination Therapy			
1	-1.05 log_{10}	Resistant to GCV / UL97	FOS + CMX001 therapy
5	-0.64 log_{10}	UL54 mutation	GCV + CMX001 therapy
Lack of a Virologic Response			
3	-0.42 log_{10}	Resistant to GCV, CDV / UL97 & UL54	

Conclusions

- Reductions in CMV viremia were seen following treatment with CMX001
- Mutations in the UL54 polymerase gene appeared to impact response to CMX001 treatment; as expected, mutations exclusively in the UL97 gene that impacted response to GCV did not affect clinical response to CMX001.

These data demonstrated that CMV viremia was reduced in these seriously ill patients with refractory CMV infection following treatment with CMX001. Based on the antiviral activity observed in these difficult to treat patients, the expanded access protocol, CMX001-350, is open to enrollment of patients with life-threatening conditions including CMV infection/disease. However, in order to have the greatest impact on clinical outcome, early intervention in the treatment of CMV infection in immunocompromised patients is important. Therefore, a randomized, placebo-controlled Phase II study of CMV pre-emption/prophylaxis in CMV R+ HSCT recipients is being conducted.