A1-2017a

Compromised Renal Function Does Not Affect the Pharmacokinetics of CMX001 in Patients with Severe Double-Stranded DNA Virus Infections

LC Trost, TK Tippin, MT Anderson and WP Painter

Chimerix, Inc., Durham, North Carolina USA

50th ICAAC
Boston, MA
September 12-15, 2010

ABSTRACT

Background

CMX001 is a lipid conjugate of cidofovir (CDV) designed to overcome the major limitations of CDV: the requirement for in-hospital intravenous administration and the potential for dose-limiting nephrotoxicity. The purpose of this report is to compare the PK and renal safety of CMX001 in patients with impaired renal function with those with normal renal function.

Methods

CMX001 has been administered orally at doses of up to 4 mg/kg to approximately 65 patients with severe double stranded DNA virus infections under Investigator held Emergency INDs. Diseases treated included disseminated AdV, CMV, EBV, HSV, JCV, and others. Treatment emergent changes in renal function were monitored by changes in relevant laboratory values. Approximately 30% of the patients were classified as renally impaired at baseline based on serum creatinine ≥ 2 mg/dL. Approximately 50% of the patients were less than 16 years old. Serial blood samples for plasma concentration analysis were obtained after the first dose. PK analysis was performed using available data from 35 patients with a sufficient number of sampling time-points.

Results

Renal Function	Adult		Pediatric	
	Mean C_{max}/Dose (ng/mL)	Mean $AUC_{0\text{-}inf}$/Dose (h*ng/mL)	Mean C_{max}/Dose (ng/mL)	Mean $AUC_{0\text{-}inf}$/Dose (h*ng/mL)
Normal	170	2160	109	1327
Compromised	125	2243	89	780

Dose normalized peak plasma concentrations (C_{max}) and systemic exposure ($AUC_{0\text{-}inf}$) to CMX001 were comparable in patients (both adult and pediatric) with and without impaired renal function at baseline. No adverse changes in renal function were attributed to CMX001; some patients had improvement in renal function during CMX001 therapy.

Conclusion

Data from renal impaired patients with severe viral infections indicate that, unlike CDV, the pharmacokinetics of CMX001 are not affected by impaired renal function. In some patients, renal function improved while on CMX001.

INTRODUCTION

CMX001 is a lipid conjugate of the acyclic nucleotide analogue cidofovir which is FDA approved as Vistide®. The clinical utility of Vistide® is limited by a requirement for administration by intravenous infusion and the risk of acute nephrotoxicity. CMX001 is readily absorbed after oral administration and is up to 1000-fold more potent *in vitro* against double stranded DNA (dsDNA) viruses compared to CDV. CMX001 also has a better distribution profile compared to CDV in that it does not selectively accumulate in kidney tissue. Thus, CMX001 offers two main advantages over CDV: it can be orally administered and has a low potential for nephrotoxicity[1]. CMX001 is currently in development for the treatment of smallpox under the animal efficacy rule and in Phase II to evaluate safety and efficacy against CMV infections in a post-transplant (solid organ and stem cell) population. A Phase II clinical trial in adenovirus is planned.

Through July 2010, CMX001 has been administered to more than 80 patients under Investigator held, Emergency INDs (EINDs) for the treatment of life-threatening double-stranded DNA virus infections. Viruses treated include vaccinia, adenovirus, CMV, HSV, EBV, BK, and JC. The majority of patients presented with a history of stem or bone marrow transplant and were not engrafted at the time CMX001 treatment was initiated. Many patients had severe renal impairment due to previous administration of Vistide® at the time CMX001 was initiated.

METHODS

CMX001 was administered once or twice weekly as 20, 50 or 100 mg tablets at approximate weight-based doses of 2, 3 or 4 mg/kg (absolute doses ranged from 20 to 350 mg). As of 31 July 2010, 1st dose PK and baseline creatinine clearance (CLcr) data were available for 46 patients.

Patient Characteristics:

Age	Pediatric (Aged ≤ 16 years), N = 20 Adult (Aged >16 to < 60 years), N = 18 Elderly (Aged ≥ 60 years), N = 8
Gender	26 Male, 20 Female
Body Weight Range	Pediatric, 4.3 to 78 kg Adult, 46 to 108 kg
Renal Replacement Therapy (RRT)	13 patients undergoing hemodialysis at initiation of CMX001
Vistide®	24 patients had received Vistide® prior to treatment with CMX001

Pharmacokinetic Determinations:

Blood samples were obtained at regular intervals from each patient following the 1st dose for analysis of CMX001 and CDV concentrations in plasma using a validated analytical method under GLP-like conditions. Non-compartmental pharmacokinetic analyses were performed using industry-standard pharmacokinetic software (WinNonlin® 5.2.1 or greater, Pharsight Corporation, Mountain View, CA) to derive estimates of standard pharmacokinetic parameters including, but not limited to, area under the plasma concentration-time curve extrapolated to infinity (AUC0-inf) and dose-normalized $AUC_{0\text{-}inf}$ ($AUC_{0\text{-}inf}$/D).

Assessment of Renal Function:

Serum creatinine was determined prior to initiation of treatment with CMX001 and periodically thereafter for the duration of treatment. Creatinine clearance was calculated using the Cockcroft-Gault equation. The 46 EIND patients in the analysis were assigned to 1 of 3 groups based on renal function using the criteria shown in the following table. Patients (n=6) with mild renal impairment (CLcr > 59 and ≤ 90; Stage 2) and patients with normal renal function undergoing dialysis to reduce fluid (n=3) were excluded resulting in 38 patients remaining for this analysis.

Group	Estimated CLcr (mL/min)	Renal Function	N
1	> 90	Normal (Stage 1)	14
2	≤ 59	Moderate and Severe Renal Impairment and Kidney Failure (Stages 3-5) Without Dialysis	13
3	≤ 59	Moderate and Severe Renal Impairment and Kidney Failure (Stages 3-5) With Dialysis	10

RESULTS*

The following table summarizes the median, dose-normalized systemic exposure ($AUC_{0\text{-}inf}$/Dose) to **CMX001** after the first dose and baseline CLcr (mL/min) in pediatric and adult EIND patients with normal renal function (Group 1) compared with moderate to severely renal impaired (stages 3-5) patients. Renal impaired patients were further divided into those not undergoing dialysis (Group 2) and those undergoing dialysis (Group 3).

Patient Group		Median Age (yr)	Median Weight (kg)	Sample Size and Gender Distribution	Median CLcr (mL/min)	Median Dose Normalized $AUC_{0\text{-}inf}$ (ng*hr/mL/D)
Pediatric (age range: 1 month to 16 years)	1	11.0	31.0	2M/5F	105.5	817.7
	2	2.4	10.7	2M/4F	37.0	731.8
	3	15	58.0	2M/1F	24.7	822.6
Adult (age range: 20 to 69 years)	1	47.0	77.8	5M/2F	146.6	1875.8
	2	63.0	63.3	4M/3F	21.2	1149.3
	3	47.0	79.0	6M/1F	25.1	3170.0

- Median systemic exposure to CMX001 in both pediatric and adult patients with renal impairment who were not receiving hemodialysis at the time of the first administration of CMX001 was similar to dose-normalized exposure in patients with normal renal function indicating that *renal impairment does not affect systemic exposure to CMX001*.
- Renal-impaired pediatric and adult patients who were undergoing dialysis at the time of the first administration of CMX001 also had similar or higher median systemic exposure to CMX001 after the first dose compared with patients with normal renal function indicating that *CMX001 exposure is not affected by hemodialysis, and therefore, it is likely that CMX001 is not removed by hemodialysis*.
- While dose-normalized exposure in adult patients receiving dialysis may appear higher than that of renal-impaired patients not receiving dialysis, this is not due to a bias attributable to differences in renal impairment as both groups have the same median creatinine clearance. It is therefore interpreted as a reflection of the variability of the CMX001 pharmacokinetics in the complex EIND patient population.

* These results were not obtained from a controlled clinical trial. The data compiled for this report were obtained from patients whose treatment with CMX001 may be ongoing and for whom final data is not available. The data were verified (QC'd) based on information provided by the investigator, however, in some cases original source data were not available.

The absence of correlation between creatinine clearance (CLcr) and dose-adjusted exposure ($AUC_{0\text{-}inf}$/D) to CMX001 is shown in the figure below. An R^2 value of < 0.001 indicates there is no proportionality between these 2 parameters. Each "x" represents one patient (n=44, $AUC_{0\text{-}inf}$ could not be estimated in 2 patients).

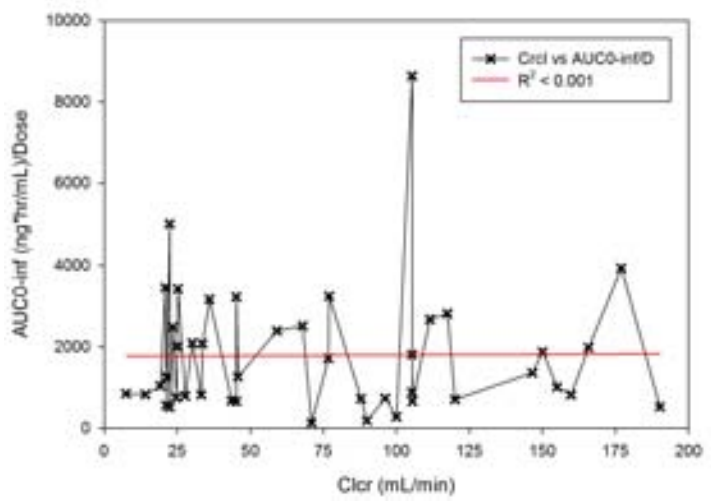
AUC0-inf (ng*hr/mL)/Dose
10000
8000
6000
4000
2000
0
0
25
50
75
100
125
150
175
200
Clcr (mL/min)
Crcl vs AUC0-inf/D
R² < 0.001

The following table summarizes the median, dose-normalized systemic exposure ($AUC_{0\text{-}inf}$/Dose) to **CDV** after the first dose of CMX001 and baseline CLcr (mL/min) in adult EIND patients with normal renal function (Group 1) compared with moderate to severely renal impaired (stages 3-5) patients. Renal impaired patients were further divided into those not undergoing dialysis (Group 2) and those undergoing dialysis (Group 3). Patients who had previously received Vistide® were excluded from the analysis. There were an insufficient number of pediatric patients who had not previously received Vistide® to include them in this analysis.

Patient Group	Median Age(yr)	Median Weight (kg)	Sample Size and Gender Distribution	Median CLcr (mL/min)	Median Dose Normalized $AUC_{0\text{-}inf}$ (ng*hr/mL/D)
1	45.3	70.7	3M/1F	135.1	236.4
2	56.0	72.0	4M/1F	21.2	1330.5
3	43.3	85.0	3M/0F	25.1	1017.3

- Median systemic exposure to CDV in adult patients with renal impairment who were not receiving hemodialysis at the time of the first administration of CMX001 was higher than the dose-normalized exposure in patients with normal renal function confirming that *renal impairment results in increased systemic exposure to CDV as described in the Vistide® Prescribing Information*.
- Renal-impaired patients who were undergoing dialysis at the time of the first administration of CMX001 also had higher dose-normalized exposure to CDV compared to patients with normal renal function, again as expected based on the Vistide® Prescribing Information.

Both baseline (pre-treatment) and post-treatment serum creatinine values were available for a subset of 25 EIND patients who did not receive hemodialysis at any time during treatment with CMX001. The figure below shows the baseline creatinine value and the most recent creatinine value obtained while the patient was receiving CMX001. The duration of treatment ranged from 1 day (single dose) to 132 days of twice weekly treatment (ongoing for a number of patients).

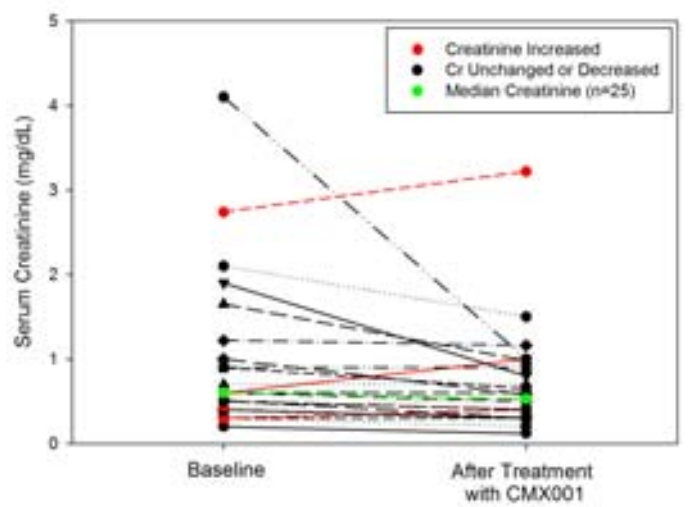
Serum Creatinine (mg/dL)
5
4
3
2
1
0
Baseline
After Treatment with CMX001
Creatinine Increased
Cr Unchanged or Decreased
Median Creatinine (n=25)

Creatinine values decreased or remained unchanged in 80% of EIND patients (20 of 25 patients). The median creatinine value decreased from 0.60 mg/dL at baseline to 0.53 mg/dL after treatment with CMX001. The increases in creatinine values observed in 5 patients during treatment were minor and not considered to be evidence of nephrotoxicity in this very compromised patient population.

CONCLUSIONS

- In separate analyses conducted in both pediatric and adult patients, moderate to severe renal impairment does not alter systemic exposure (AUC) to CMX001 compared to patients with normal renal function.
- CMX001 exposure is not affected by hemodialysis.
- Renal function improved or remained unchanged in the majority of EIND patients during administration of CMX001 supporting the lack of nephrotoxicity associated with CMX001.

ACKNOWLEDGEMENTS

The authors wish to acknowledge our colleagues, Dr. Herve Mommeja-Marin and Timothy King, for critical evaluation of the poster and assistance with data analysis.

REFERENCES

1. Painter, R.G., Trost, C.L., Lampert, M.B., Almond, R.M., Buller, M., Kern, E., Painter, G.P., Robertson, A.T., Frazer, N., O'Mahony, R (2008) CMX001. Anti-smallpox agent, anti-cytomegalovirus agent, viral polymerase inhibitor. *Drugs Fut.* 2008, **33**(8): 655.

CHIMERIX, INC.